Safety and Efficacy of VP-102 (cantharidin, 0.7% w/v) in Molluscum Contagiosum by Lesion Body Region by Day in Pooled Phase 3 Trials

Lawrence F. Eichenfield[1], Albert Yan[2], Pearl Kwong[3], Mercedes E. Gonzalez[4], Pieter d'Arnaud[5], Patrick Burnett[6], Melissa Olivadoti[6]

[1]UC San Diego and Rady Children's Hospital, San Diego, CA, [2]Children's Hospital of Philadelphia, Philadelphia, PA, [3]Solutions Through Advanced Research, Jacksonville, FL, [4]Skin Research Institute, Coral Gables, FL, [5]Instat Consulting, Inc., Chatham, NJ, [6]Verrica Pharmaceuticals Inc., West Chester, PA

VP-102

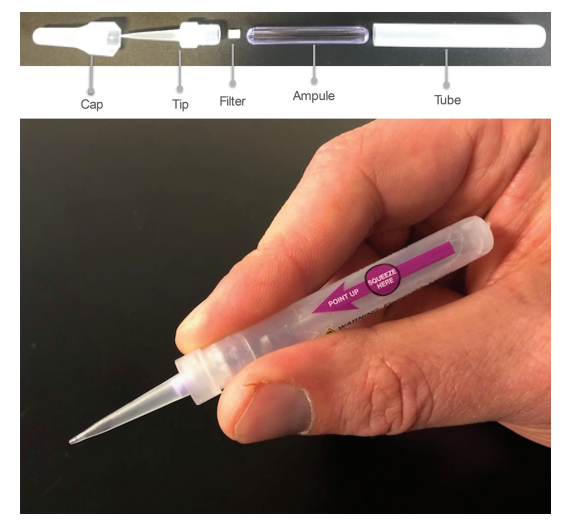
Cap
Tip
Filter
Ampule
Tube

VP-102 is a proprietary drug-device combination that is not approved by the FDA. Photo is for representative purposes only.

BACKGROUND

- VP-102 is a proprietary drug-device combination product containing a controlled formulation of cantharidin (0.7% w/v) that has been investigated in two Phase 3 trials for the treatment of molluscum contagiosum.
- Anatomical and epidermal differences across distinct areas of the body could lead to variations in efficacy and safety by body region.
- The objective of this exploratory analysis was to determine the safety and efficacy of VP-102 by analyzing pooled data segmented by regions of the body by visit/day.

METHODS

- Segmentation of lesions by body region at baseline included the following areas:
 - Head/Neck
 - Chest/Abdomen
 - Back/Buttocks
 - Groin
 - Upper Extremities
 - Lower Extremities
- Lesion counts by body region were obtained at each visit (Days 1, 21, 42, 63, and 84). Efficacy was measured by complete clearance of baseline and new lesions in the identified region.
- Subjects could present with multiple body regions at baseline. Lesions occurring in new regions after baseline were not tracked in these analyses.
- Safety analyses were conducted for subjects with lesions treated in the identified body region during or after treatment Visit 1 and before treatment Visit 2.
- The delineations of body areas and determination of treatability of lesions located within 10 mm of mucosal openings were made by the investigators.

MOLLUSCUM BASELINE CHARACTERISTICS

Body Regions With Lesions at Baseline (ITT Population)*

	VP-102 (n=310)	Vehicle (n=218)
Total Number of Body Regions with Lesions at Baseline*	729	557
Total Number of Body Regions with Lesions at Baseline by Subject*		
n	309	217
Mean	2.36	2.57
SD	1.197	1.304
Median	2.00	2.00
Min-Max	1.0–6.0	1.0–6.0
Number of Subjects with Lesions at Baseline - No(%)		
Head/Neck	77 (24.8)	53 (24.3)
Chest/Abdomen	142 (45.8)	118 (54.1)
Back/Buttocks	117 (37.7)	91 (41.7)
Groin	28 (9.0)	25 (11.5)
Upper Extremities	179 (57.7)	131 (60.1)
Lower Extremities	186 (60.0)	141 (64.7)

There were no significant differences in mean number of lesions or total number of body regions at baseline between the VP-102 and vehicle groups (all p>0.05).

* Only those subjects who were assessed for lesions in all six body regions at baseline were considered.

SAFETY

Incidence of Application Site TEAEs by Body Location in VP-102-Treated Subjects (Safety Population)

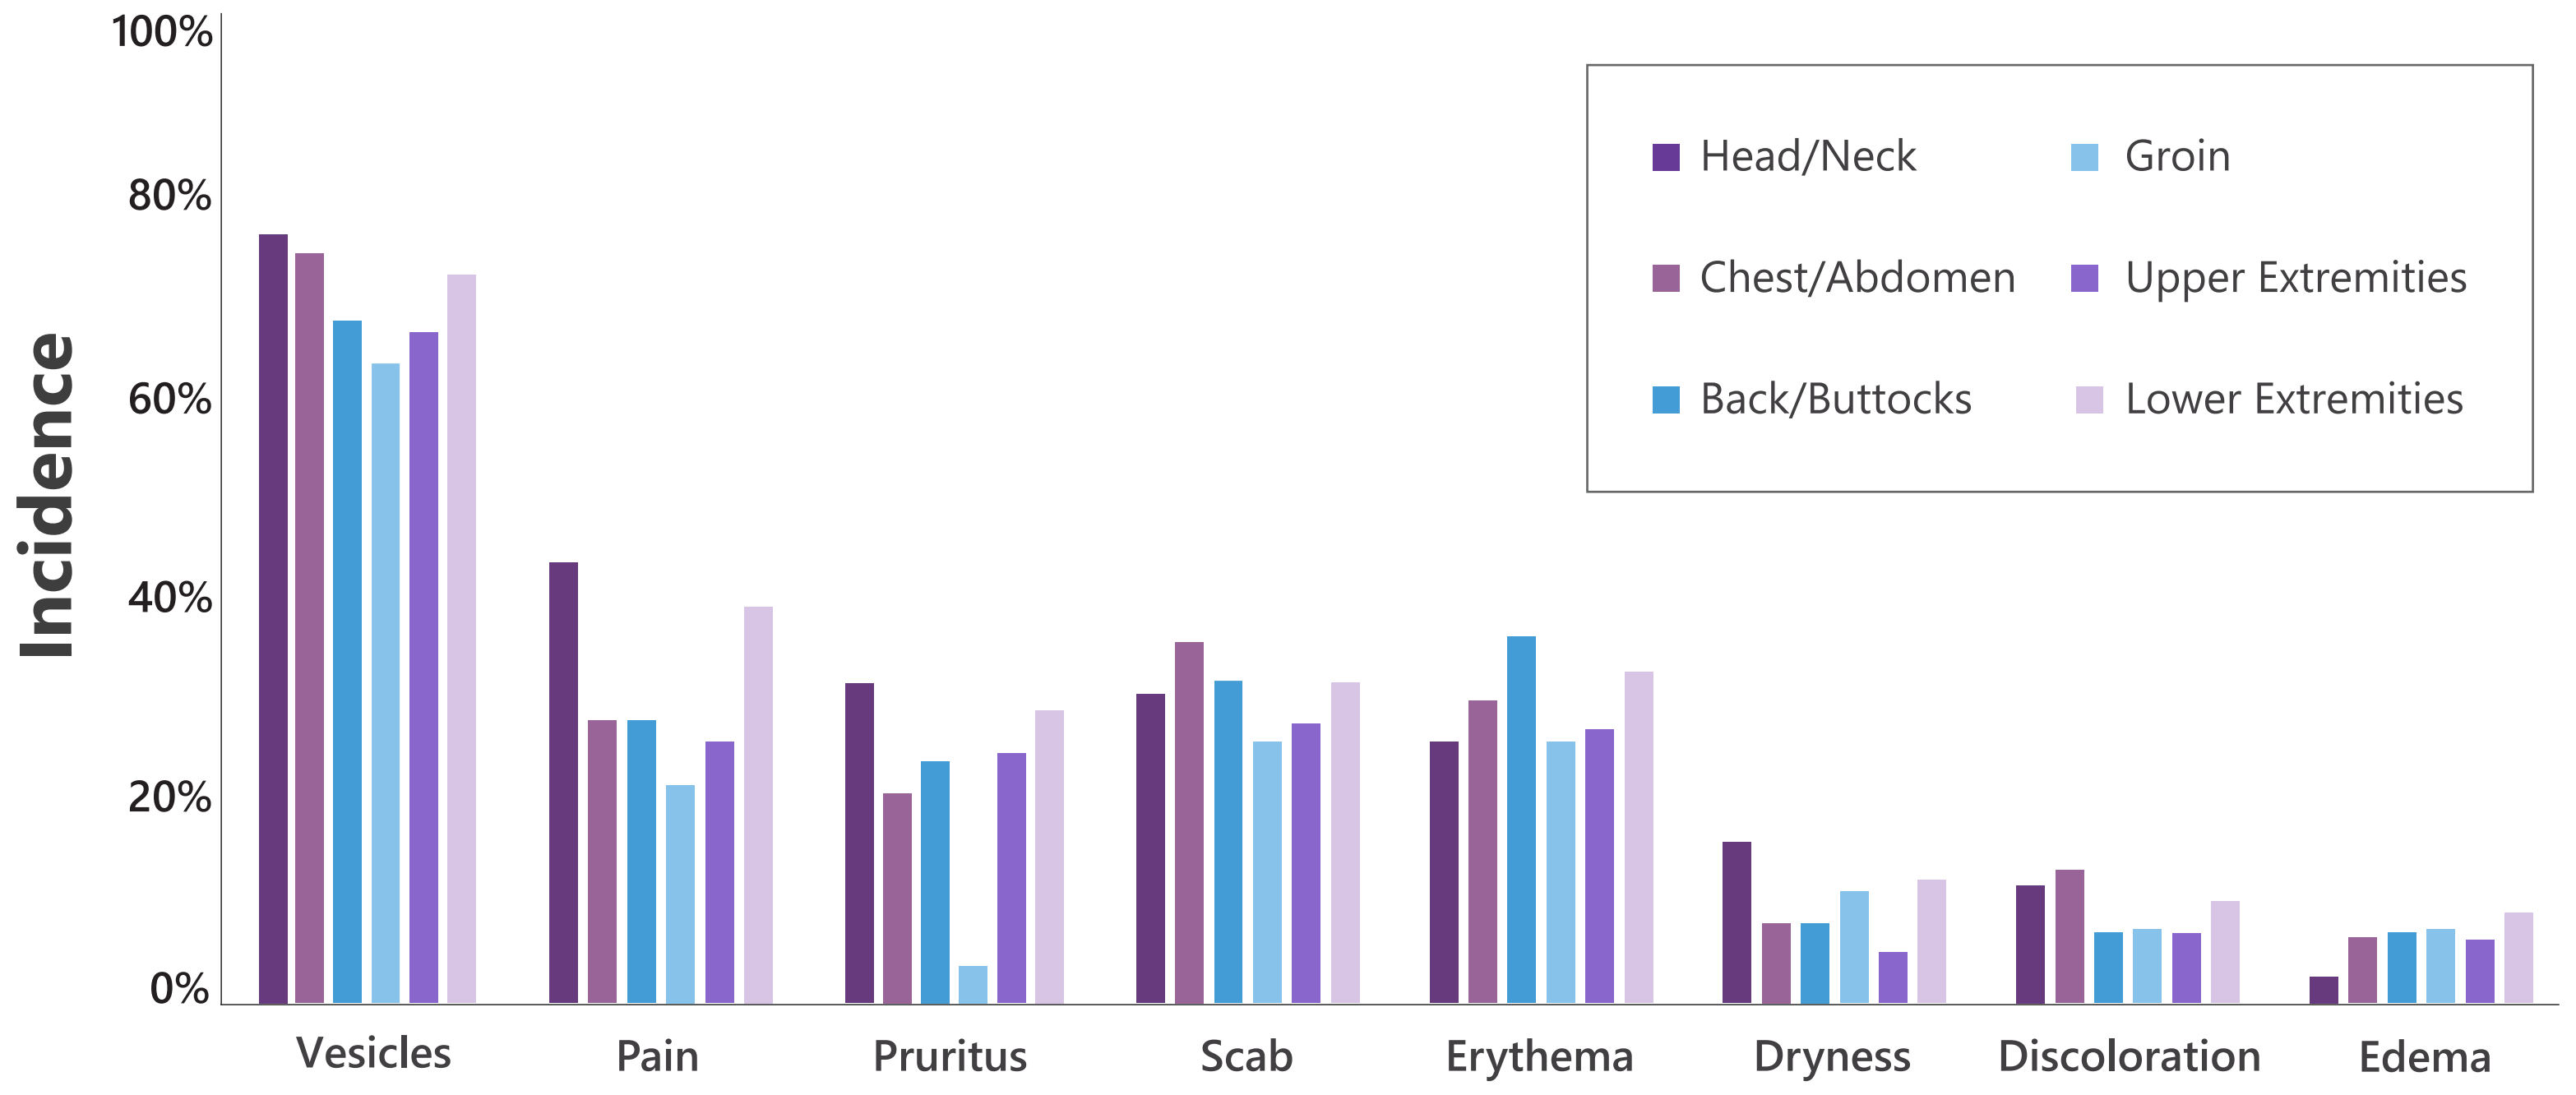
Incidence
100%
80%
60%
40%
20%
0%
Head/Neck
Chest/Abdomen
Back/Buttocks
Groin
Upper Extremities
Lower Extremities
Vesicles
Pain
Pruritus
Scab
Erythema
Dryness
Discoloration
Edema

EFFICACY

Subjects with Complete Lesion Clearance by Body Region by Visit/Day (ITT Population)

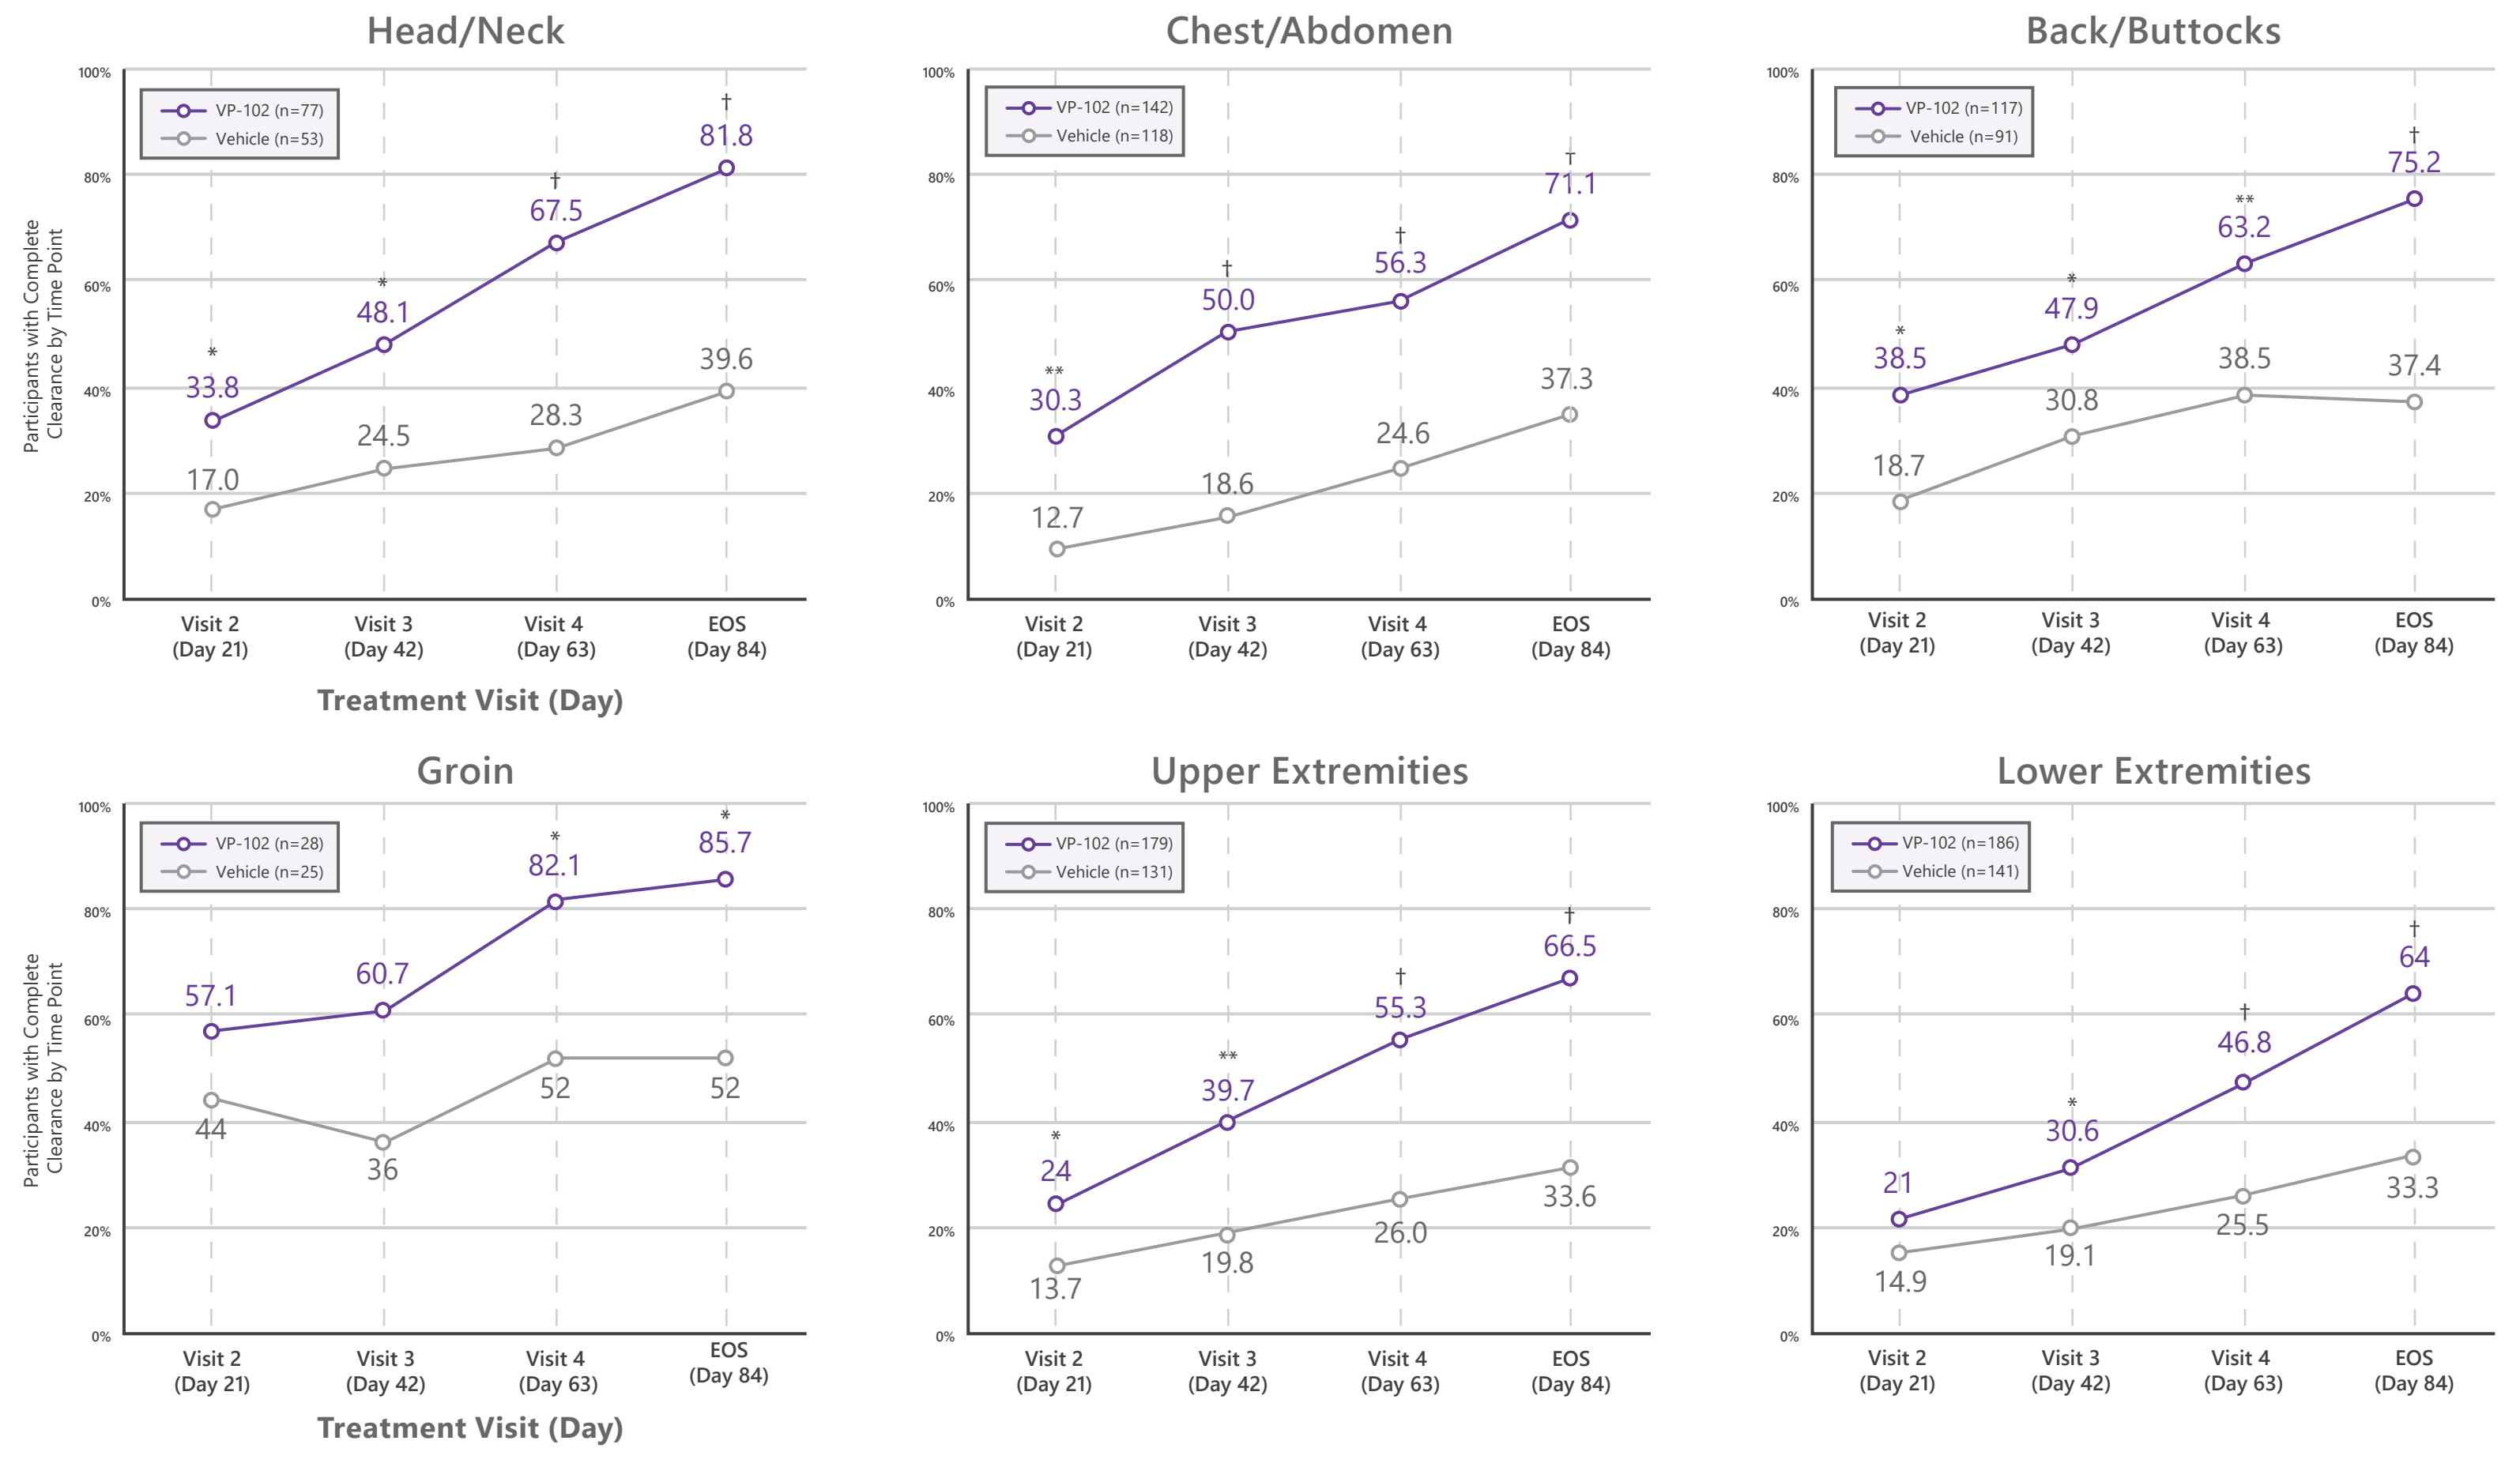
Head/Neck
VP-102 (n=77)
Vehicle (n=53)
33.8
48.1
67.5
81.8
17.0
24.5
28.3
39.6
Chest/Abdomen
VP-102 (n=142)
Vehicle (n=118)
30.3
50.0
56.3
71.1
12.7
18.6
24.6
37.3
Back/Buttocks
VP-102 (n=117)
Vehicle (n=91)
38.5
47.9
63.2
75.2
18.7
30.8
38.5
37.4
Groin
VP-102 (n=28)
Vehicle (n=25)
57.1
60.7
82.1
85.7
44
36
52
52
Upper Extremities
VP-102 (n=179)
Vehicle (n=131)
24
39.7
55.3
66.5
13.7
19.8
26.0
33.6
Lower Extremities
VP-102 (n=186)
Vehicle (n=141)
21
30.6
46.8
64
14.9
19.1
25.5
33.3
Participants with Complete Clearance by Time Point
Visit 2 (Day 21)
Visit 3 (Day 42)
Visit 4 (Day 63)
EOS (Day 84)
Treatment Visit (Day)

* $p \leq 0.05$, ** $p \leq 0.001$, † $p \leq 0.0001$

EOS=end of study.

CONCLUSIONS

- In all body regions, treatment with VP-102 resulted in a statistically signficantly greater percentage of subjects with clearance of baseline and new molluscum lesions compared to vehicle.
- Different body regions may require different numbers of treatments.
- The incidence of adverse events was similar in the six body regions and visits.
- Further studies are needed to make strong conclusions about use in the groin and face/neck region due to lower number of subjects in these groups.

Disclosures

This study was sponsored by Verrica Pharmaceuticals Inc. Editorial support was provided by Versant Learning Solutions and funded by Verrica Pharmaceuticals Inc.