Phase 2 Safety and Efficacy of VP-102, a Drug-Device Combination Product Containing Cantharidin (0.7% w/v), for the Treatment of External Genital Warts (CARE-1)

Guenthner, S[1]; McFalda, W[2]; Tate, M[3]; Eads, K[1]; d'Arnaud, P[4]; Rieger, J[5]; Glover, D[5]; Willson, C[5]; Rosen, T[6]; Olivadoti, M[5]

[1]The Indiana Clinical Trials Center; [2]Clarkston Skin Research; [3]DelRicht Research, Tulsa, OK; [4]Instat Consulting Inc, Chatham, NJ; [5]Verrica Pharmaceuticals Inc., West Chester, PA; [6]Baylor College of Medicine, Houston, TX

VP-102

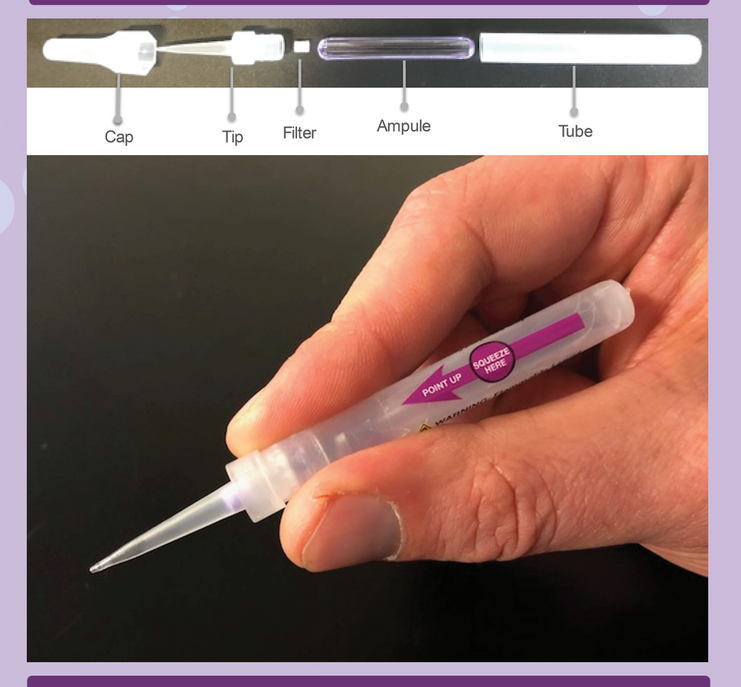
Cap
Tip
Filter
Ampule
Tube

VP-102 is a proprietary drug-device combination that is not approved by FDA. Photo is for representative purposes only.

INTRODUCTION

- External genital warts (EGW) are caused by the human papilloma virus, which is spread through direct skin-to-skin contact.[1]
- Approximately 1% of people in the US have EGW.[2]
- Cantharidin has been used to treat EGW for decades,[3] however the treatment remains unapproved by the FDA, with no reliable or controlled source available.

METHODS

- This Phase 2, double-blind, vehicle-controlled trial included two parts (A and B, see Figure 1).
- The number of EGW was required to be between 2 and 30 and located within the medial thigh, supra-pubic area, and/or perianal area.
- The primary efficacy endpoint was measured by the percentage of subjects who had complete clearance of all baseline and new EGW lesions at Day 84 (end of treatment visit, EOT).
- Safety was assessed via related local skin reactions (LSRs) and adverse events (AEs).

Figure 1. Study design for CARE-1 Phase 2 clinical trial. There were 2 parts of the study: A and B.

Part A

Treatment Group	VP-102	Vehicle
5 : 1 Randomization		
Exposure*	2, 6, or 24 hours	

Part B

Treatment Group	VP-102	Vehicle
3 : 2 Randomization		
Exposure*	6 or 24 hours	

Day 1 Visit 1 → Day 3 Safety Evaluation → Day 21 Visit 2 → Day 42 Visit 3 → Day 63 Visit 4 → Day 84 Visit EOT

EOT=end of treatment. * Duration of exposure prior to washing off the medication.

RESULTS

Table 1. CARE-1 subject demographics and EGW characteristics.

	VP-102 6-hours (n=30)	Vehicle 6-hours (n=24)	VP-102 24-hours (n=27)	Vehicle 24-hours (n=18)
Age				
Mean (SD)	38.93 (9.9)	35.83 (7.8)	34.33 (7.1)	33.83 (6.3)
Min, Max	26, 59	26, 58	25, 53	25, 43
Gender, n (%)				
Male	17 (56.7)	14 (58.3)	15 (55.6)	11 (61.1)
Female	13 (43.3)	10 (41.7)	12 (44.4)	7 (38.9)
Race, n (%)				
White	24 (80.0)	13 (54.2)	24 (88.9)	12 (66.7)
Black or African American	6 (20.0)	8 (33.3)	2 (7.4)	6 (33.3)
American Indian or Alaska Native	0 (0)	1 (4.2)	0 (0)	0 (0)
Other	0 (0)	2 (8.3)	1 (3.7)	0 (0)
Ethnicity, n (%)				
Hispanic or Latino	6 (20.0)	1 (4.2)	2 (7.4)	5 (27.8)
Not Hispanic or Latino	24 (80.0)	23 (95.8)	25 (92.6)	13 (72.2)
Duration of Warts, No. (%)				
<1 year	15 (50.0)	12 (50.0)	13 (48.1)	9 (50.0)
1–2 years	3 (10)	1 (4.2)	2 (7.4)	0 (0.0)
>2–5 years	4 (13.3)	5 (20.8)	8 (29.6)	3 (16.7)
>5 years	8 (26.7)	6 (25.0)	3 (11.1)	6 (33.3)
Wart Count				
Mean	8.5	6.71	9.48	7.56
SD	7.3	5.5	6.2	6.8
Median	6	5	9	4.5
Min, Max	2, 30	2, 26	2, 25	2, 28
Prior Wart Treatment, No. %				
Yes	17 (56.7)	13 (54.2)	14 (51.9)	9 (50)

Pooled data from the 6- and 24-hours groups from Part A and Part B; ITT Population.

Figure 2. Percentage of subjects with complete clearance of all baseline and new treatable EGW by visit.

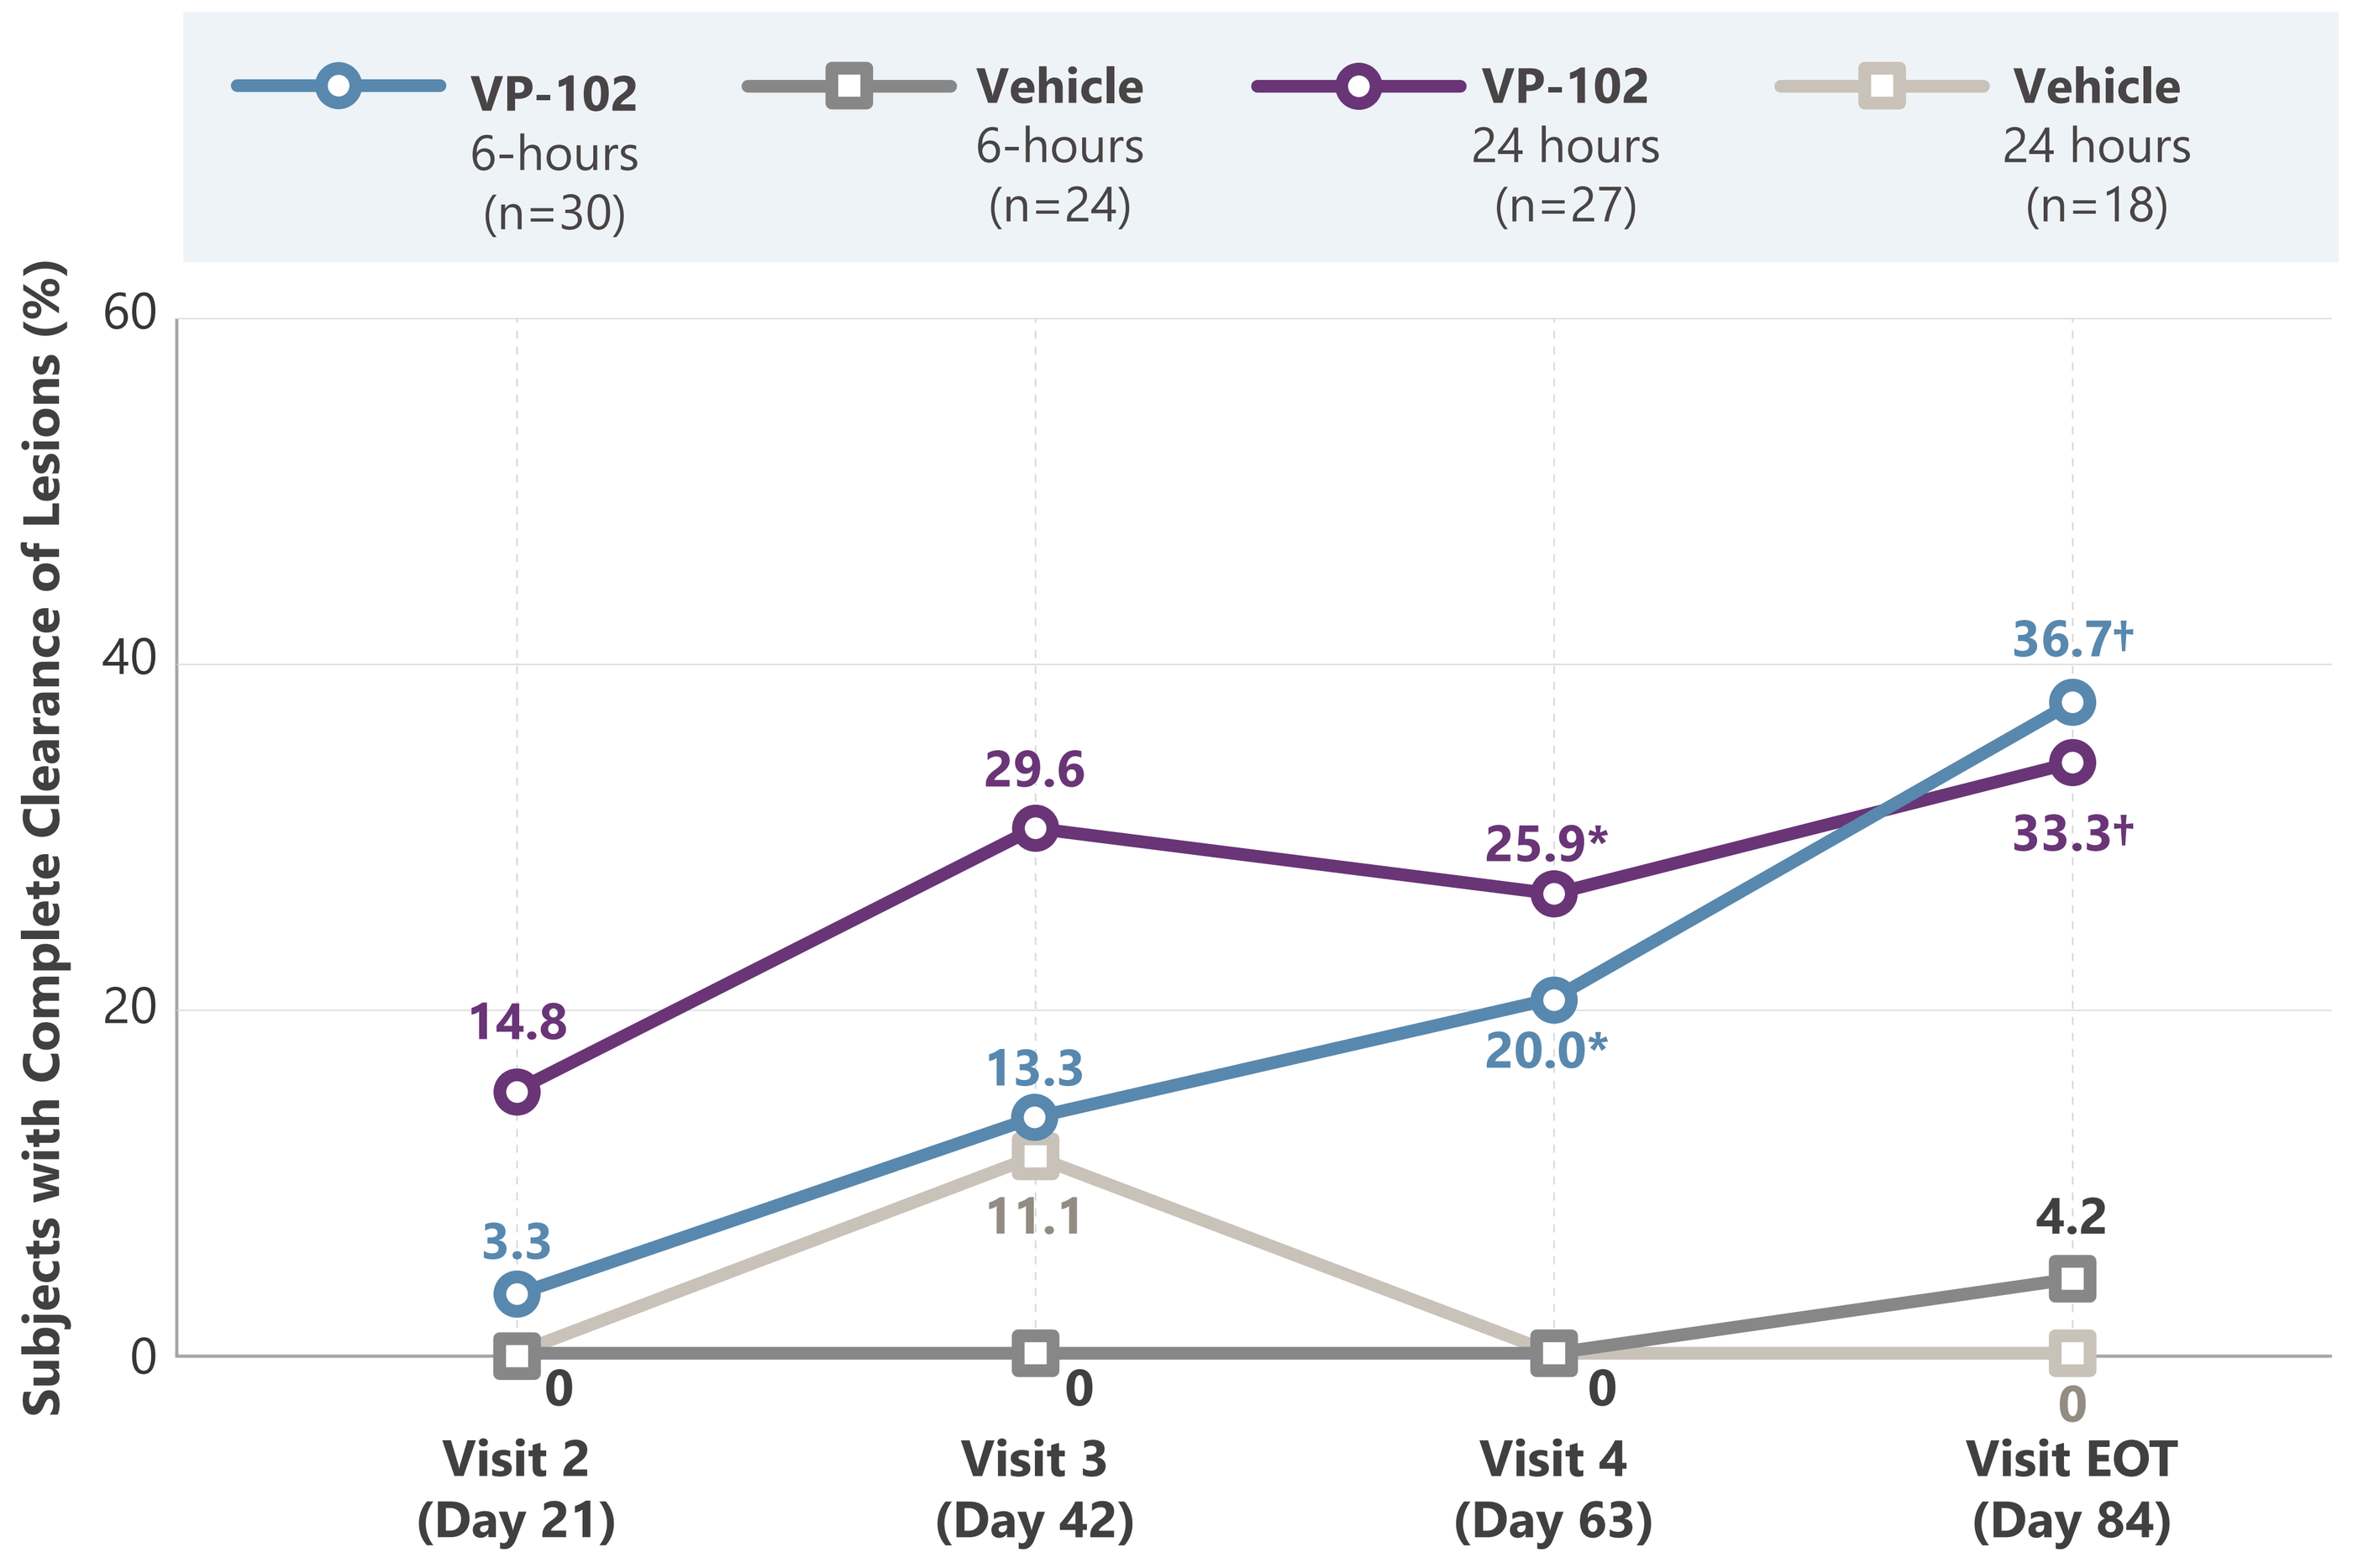
VP-102 6-hours (n=30)
Vehicle 6-hours (n=24)
VP-102 24 hours (n=27)
Vehicle 24 hours (n=18)
Subjects with Complete Clearance of Lesions (%)
60
40
20
0
14.8
29.6
25.9*
33.3†
3.3
13.3
20.0*
36.7†
11.1
4.2
0
0
0
0
Visit 2 (Day 21)
Visit 3 (Day 42)
Visit 4 (Day 63)
Visit EOT (Day 84)

* $p \leq 0.05$ (VP-102 regimen vs. vehicle)
† $p \leq 0.01$ (VP-102 regimen vs. vehicle)

Pooled data from the 6- and 24-hours groups from Part A and Part B; ITT Population.

SAFETY

Table 2. Treatment emergent adverse events related to study drug (≥5%, Safety Population).

TEAEs, N (%)	VP-102 6-hours (n=29)	Vehicle 6-hours (n=22)	VP-102 24-hours (n=28)	Vehicle 24-hours (n=20)
Subjects reporting at least one TEAE	29 (100.0)	8 (36.4)	28 (100.0)	6 (30.0)
Application site vesicles	25 (86.2)	0 (0.0)	26 (92.9)	1 (5.0)
Application site pain	20 (69.0)	3 (13.6)	19 (67.9)	4 (20.0)
Application site erythema	14 (48.3)	3 (13.6)	19 (67.9)	1 (5.0)
Application site pruritus	14 (48.3)	5 (22.7)	10 (35.7)	1 (5.0)
Application site scab	13 (44.8)	1 (4.5)	14 (50.0)	0 (0.0)
Application site discoloration	7 (24.1)	4 (18.2)	6 (21.4)	0 (0.0)
Application site dryness	7 (24.1)	2 (9.1)	6 (21.4)	1 (5.0)
Application site erosion	6 (20.7)	0 (0.0)	7 (25.0)	0 (0.0)
Application site edema	3 (10.3)	1 (4.5)	7 (25.0)	1 (5.0)
Application site exfoliation	3 (10.3)	2 (9.1)	5 (17.9)	0 (0.0)

No serious adverse events occurred that were deemed by the investigator to be related to the treatment.

CONCLUSIONS

- This Phase 2 study demonstrated the safety and efficacy of VP-102 in the treatment of EGW.
 - The 6- and 24-hour duration periods had similar outcomes for efficacy and safety.
- Treatment with VP-102 resulted in statistically significantly higher complete clearance of all EGW at day 63 and 84 compared to vehicle.
- TEAEs in the VP-102 group were mostly mild to moderate and expected due to the pharmacodynamic action of cantharidin, the active ingredient in VP-102. TEAEs were similar across different exposure times.
- VP-102 is well-tolerated for the treatment of EGW, as no subjects discontinued due to AEs and there were no serious AEs reported due to study drug.

References

1. Recanati MA, et. Al. *Clin Exp Obstet Gynecol*. 2018;45(3):383-386.
2. CDC. Genital HPV Infection Fact Sheet. Accessed February 8, 2019. Available at: https://www.cdc.gov/std/hpv/stdfact-hpv.htm.
3. Epstein WL, Kligman AM. *AMA Arch Derm*. 1958;77(5):508-511

Funding Statement

Verrica Pharmaceuticals Inc. provided funding for the study and for the creation of the poster by Versant Learning Solutions.