The Hidden Impact of Molluscum Contagiosum: Survey of Caregivers' Experiences with Diagnosis and Management

Pearl Kwong[1], Adelaide Hebert[2], Collette Utley[3], Melissa Olivadoti[4]

[1]Solutions Through Advanced Research, Jacksonville, FL; [2] UTHealth McGovern Medical School, Houston, TX; [3]Gold Skin Care Center, Nashville, TN; [4]Verrica Pharmaceuticals Inc., West Chester, PA

INTRODUCTION

- Molluscum contagiosum ("molluscum") is a common pediatric viral skin infection that is considered benign and self-limiting. However, it may affect the quality of life in patients and their caregivers dealing with the disease.
- The experience of caregiving for a child with molluscum is largely unknown, including the emotional burden, effort for diagnosis, and treatment modalities used for molluscum.

METHODS

- Parents, caregivers, and legal guardians (ages 18+, 80% female, 20% male) of children diagnosed with molluscum in the past 4 years (ages 3–16 years) were recruited from a panel of previously recruited respondents to answer a 15-minute paid online survey.
- Question topics included the type of health care professionals (HCPs) consulted about diagnosis, treatment (both in the clinic and at home), and the impact of the virus on the caregiver and the child.
- The margin of error at the 95% confidence interval for the total sample was ± 7.7%.

RESULTS

Table 1. Survey Respondent Demographics and Characteristics

	Respondents (N=154)
Household Information	
Median number of people in the home	4
Median age of children in the home in years	8
Household size – No. (%)	
2-3 People	44 (28)
4-5 People	94 (61)
6+ People	16 (10)
Age of Children in the Home – No. (%)	
Under 3 years	48 (31)
4-8 years	80 (52)
9-12 years	77 (50)
13-16 years	67 (43)
Caregiver Gender – No. (%)	
Male	31 (20)
Female	123 (80)
Molluscum Status – No. (%)	
Child currently has molluscum	116 (75)
Child had molluscum in the last 4 years	39 (25)
Spread of molluscum from one child to another in the home*	65 (60)
Ethnicity – No. (%)	
White/Caucasian	131 (85)
Black/African American	16 (10)
Asian	6 (4)
Native Hawaiian/Pacific Islander	1 (1)
American Indian/Alaskan Native	2 (1)
Other	3 (2)
Homestead information – No. (%)	
City	79 (51)
Suburb	65 (42)
Small town/rural	10 (7)

* In homes of >1 child, n=108.

Figure 1. "I worried about what people thought about our family having a child with molluscum."

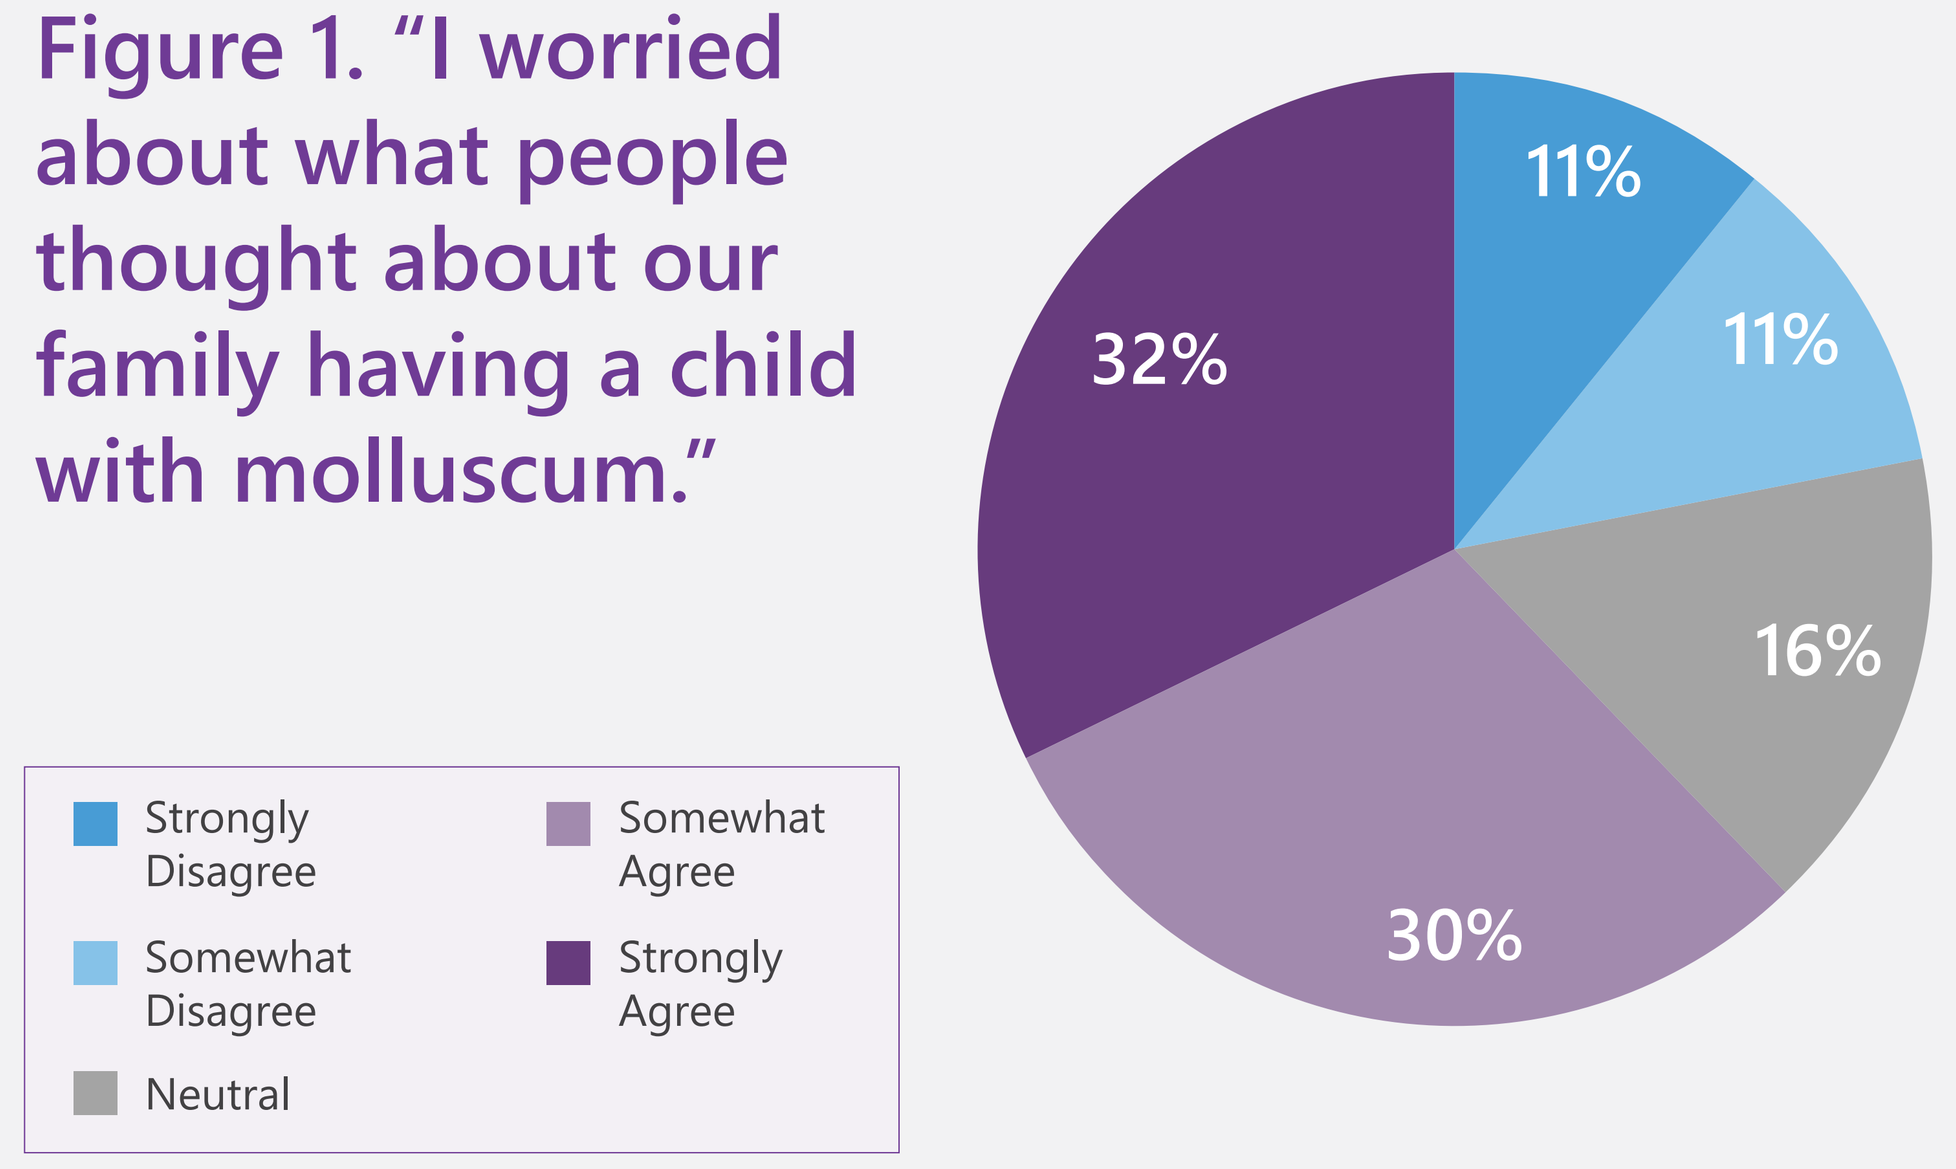
Strongly Disagree
Somewhat Disagree
Neutral
Somewhat Agree
Strongly Agree
11%
11%
16%
30%
32%

Figure 2. Types of HCPs Consulted About Molluscum Contagiosum

The average number of HCPs consulted per caregiver was 1.95.

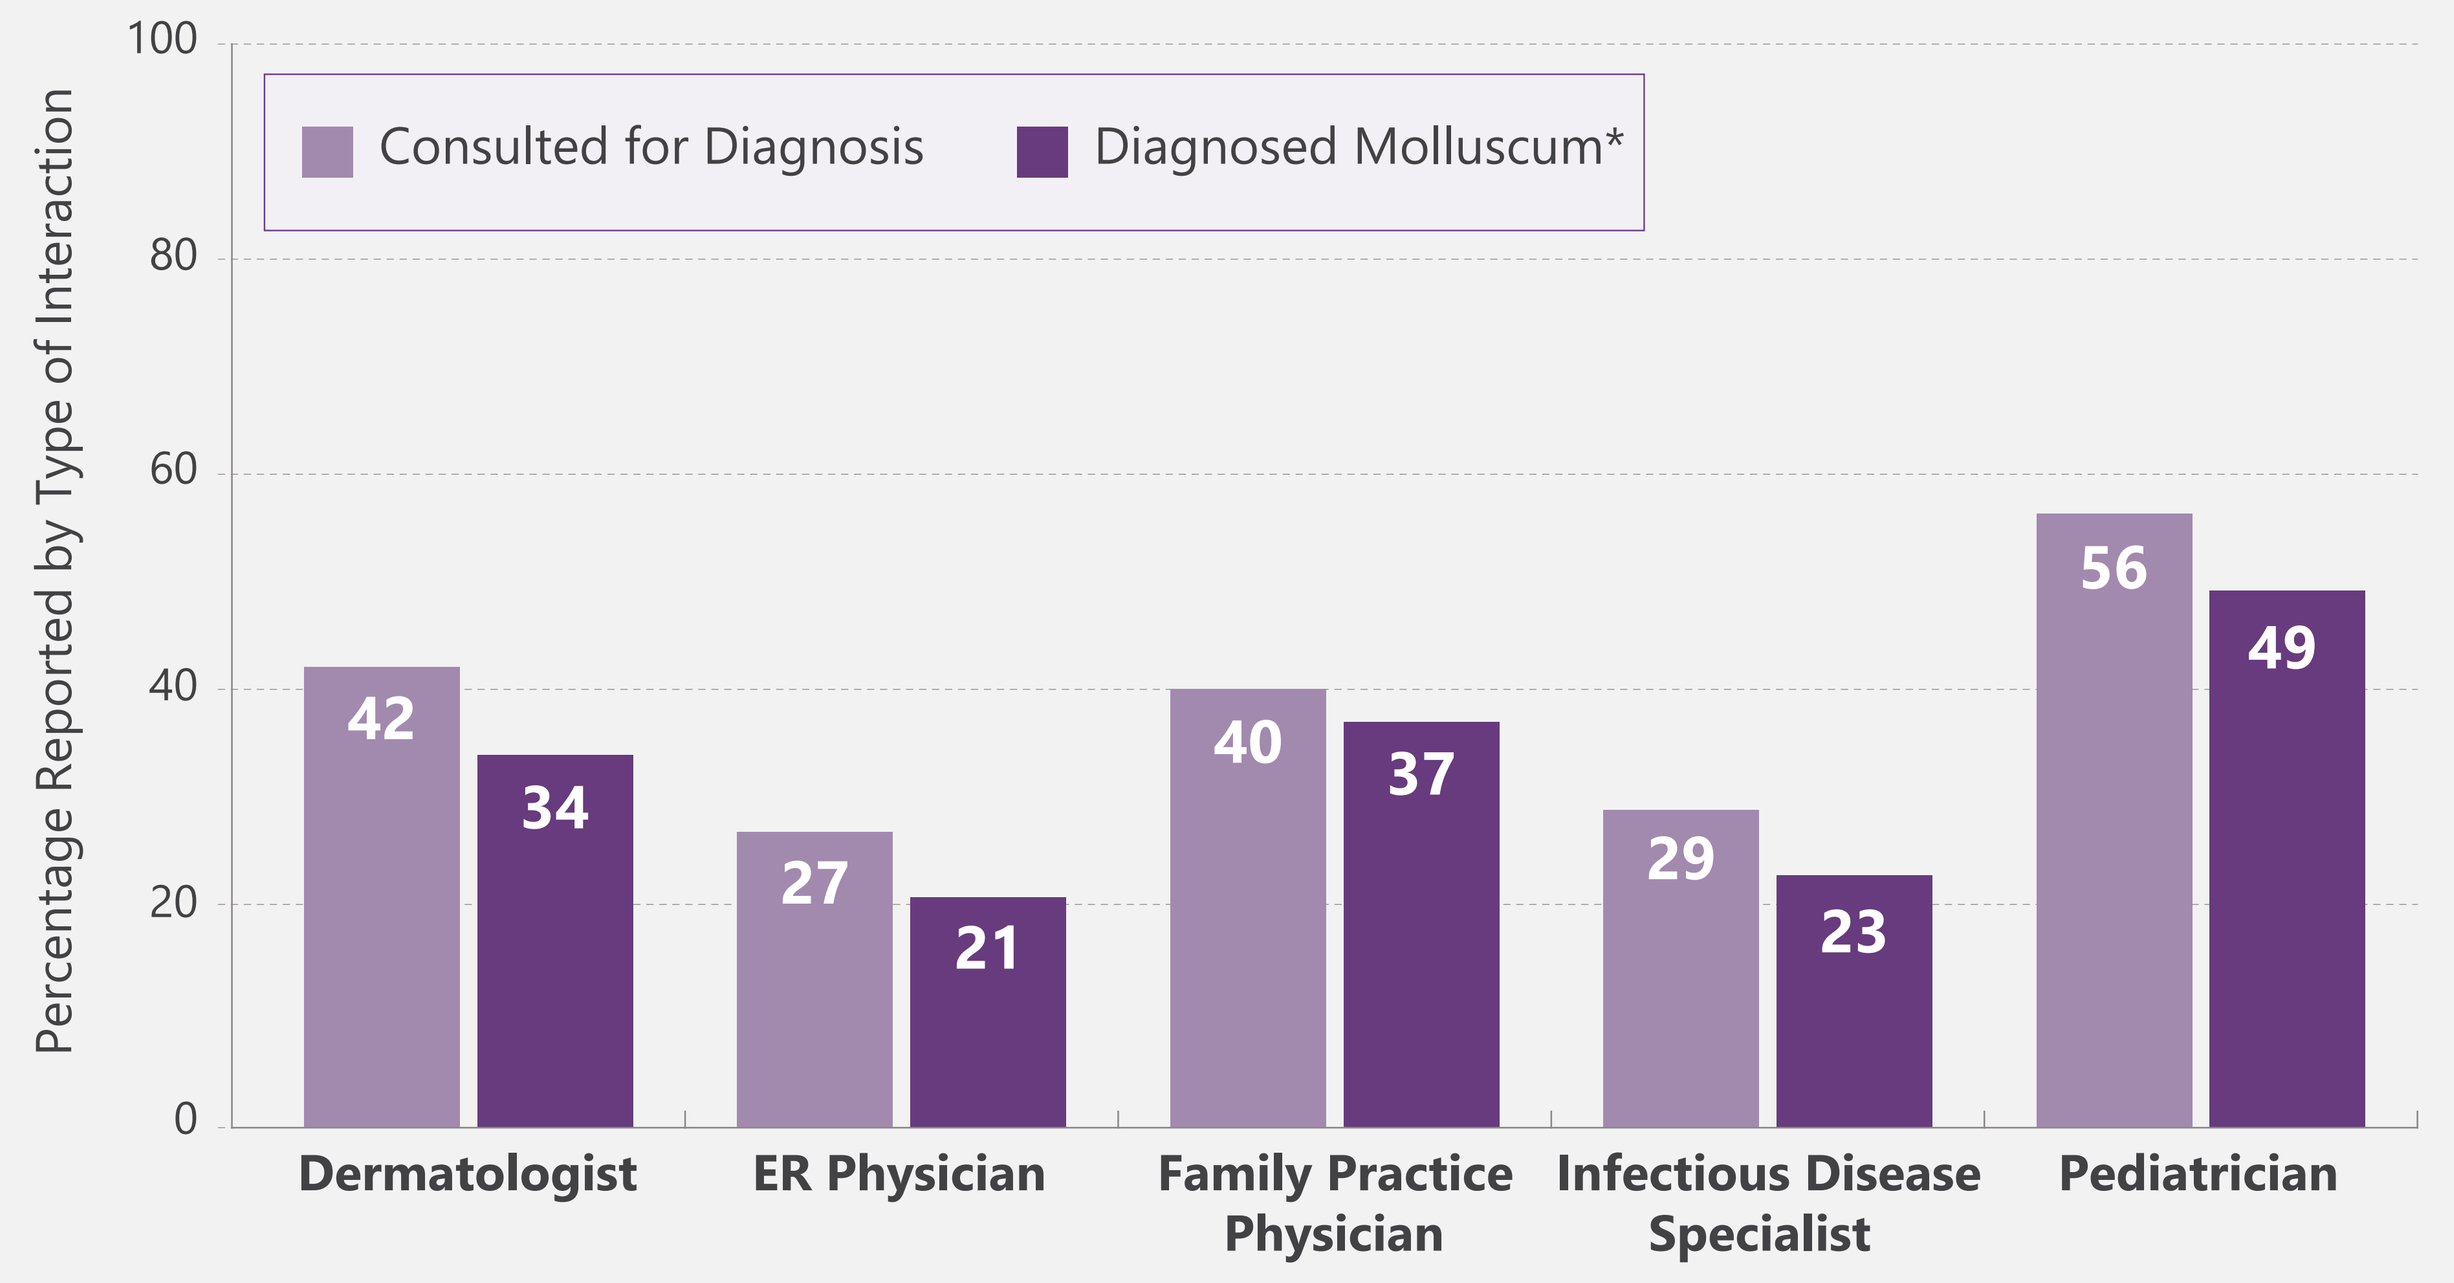
Percentage Reported by Type of Interaction
100
80
60
40
20
0
Consulted for Diagnosis
Diagnosed Molluscum*
42
34
27
21
40
37
29
23
56
49
Dermatologist
ER Physician
Family Practice Physician
Infectious Disease Specialist
Pediatrician

* Caregivers may have sought and received a diagnosis of molluscum for their child from more than one HCP type.

Figure 3. HCP Treatment Recommendations

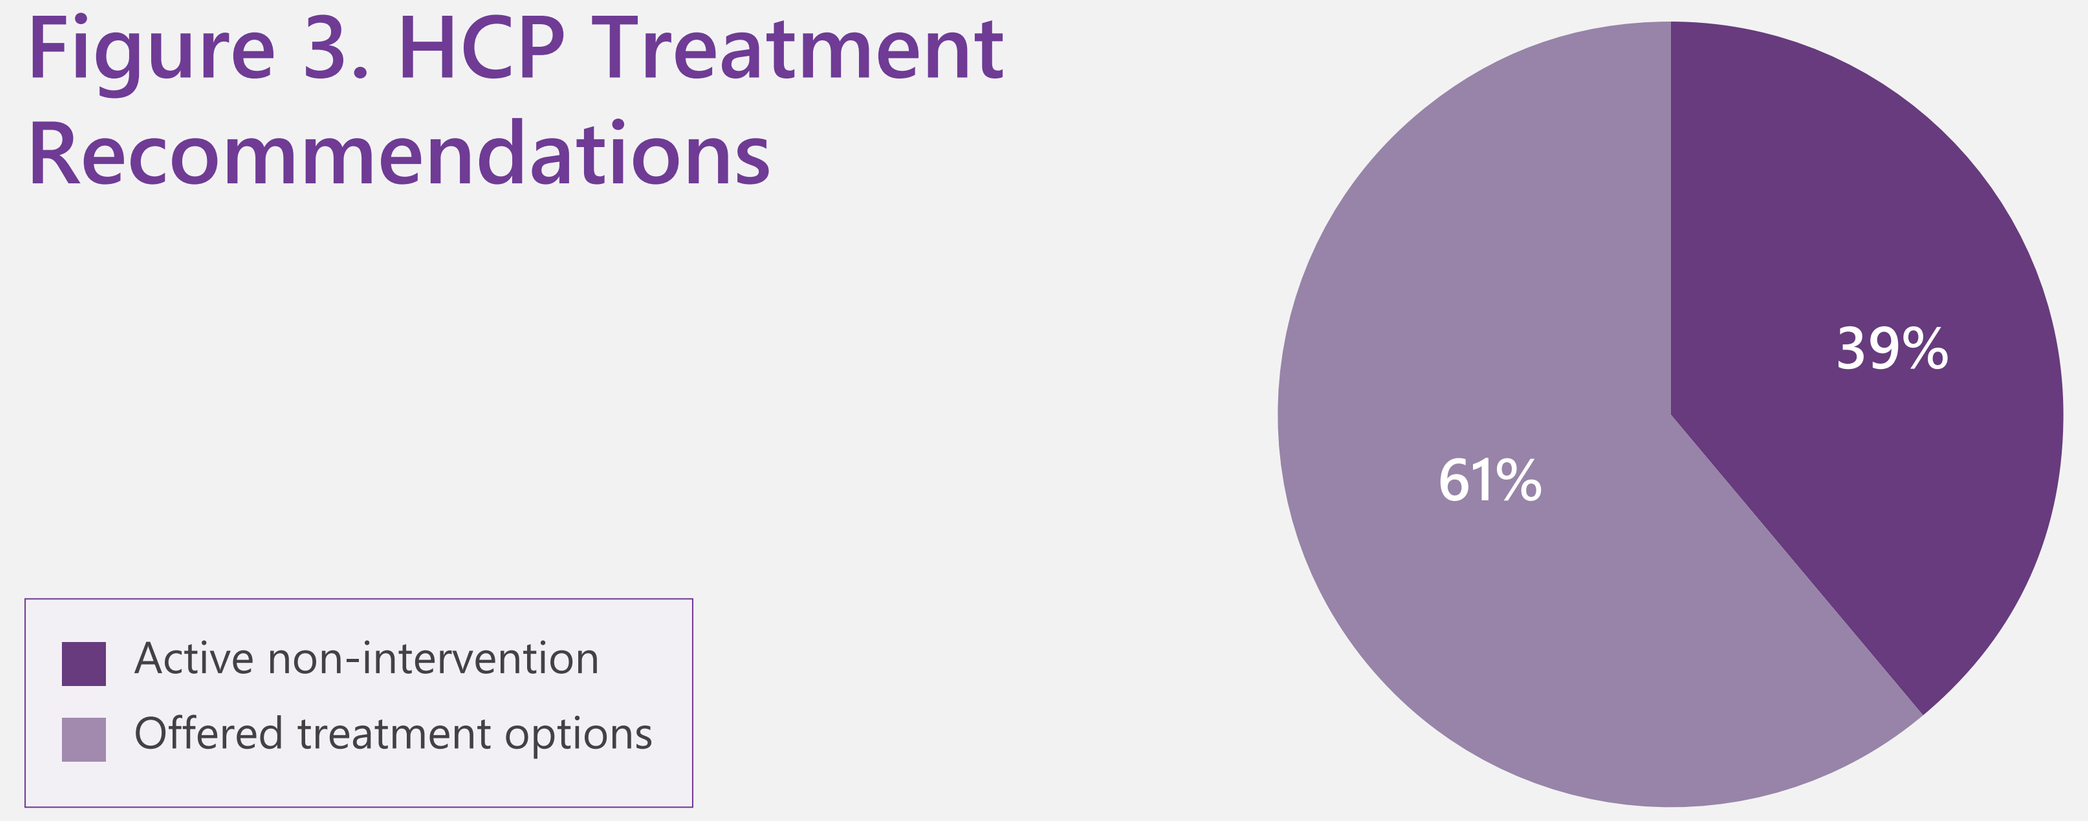
Active non-intervention
Offered treatment options
39%
61%

Figure 4. "Which of the following do you remember the doctor telling you?"*

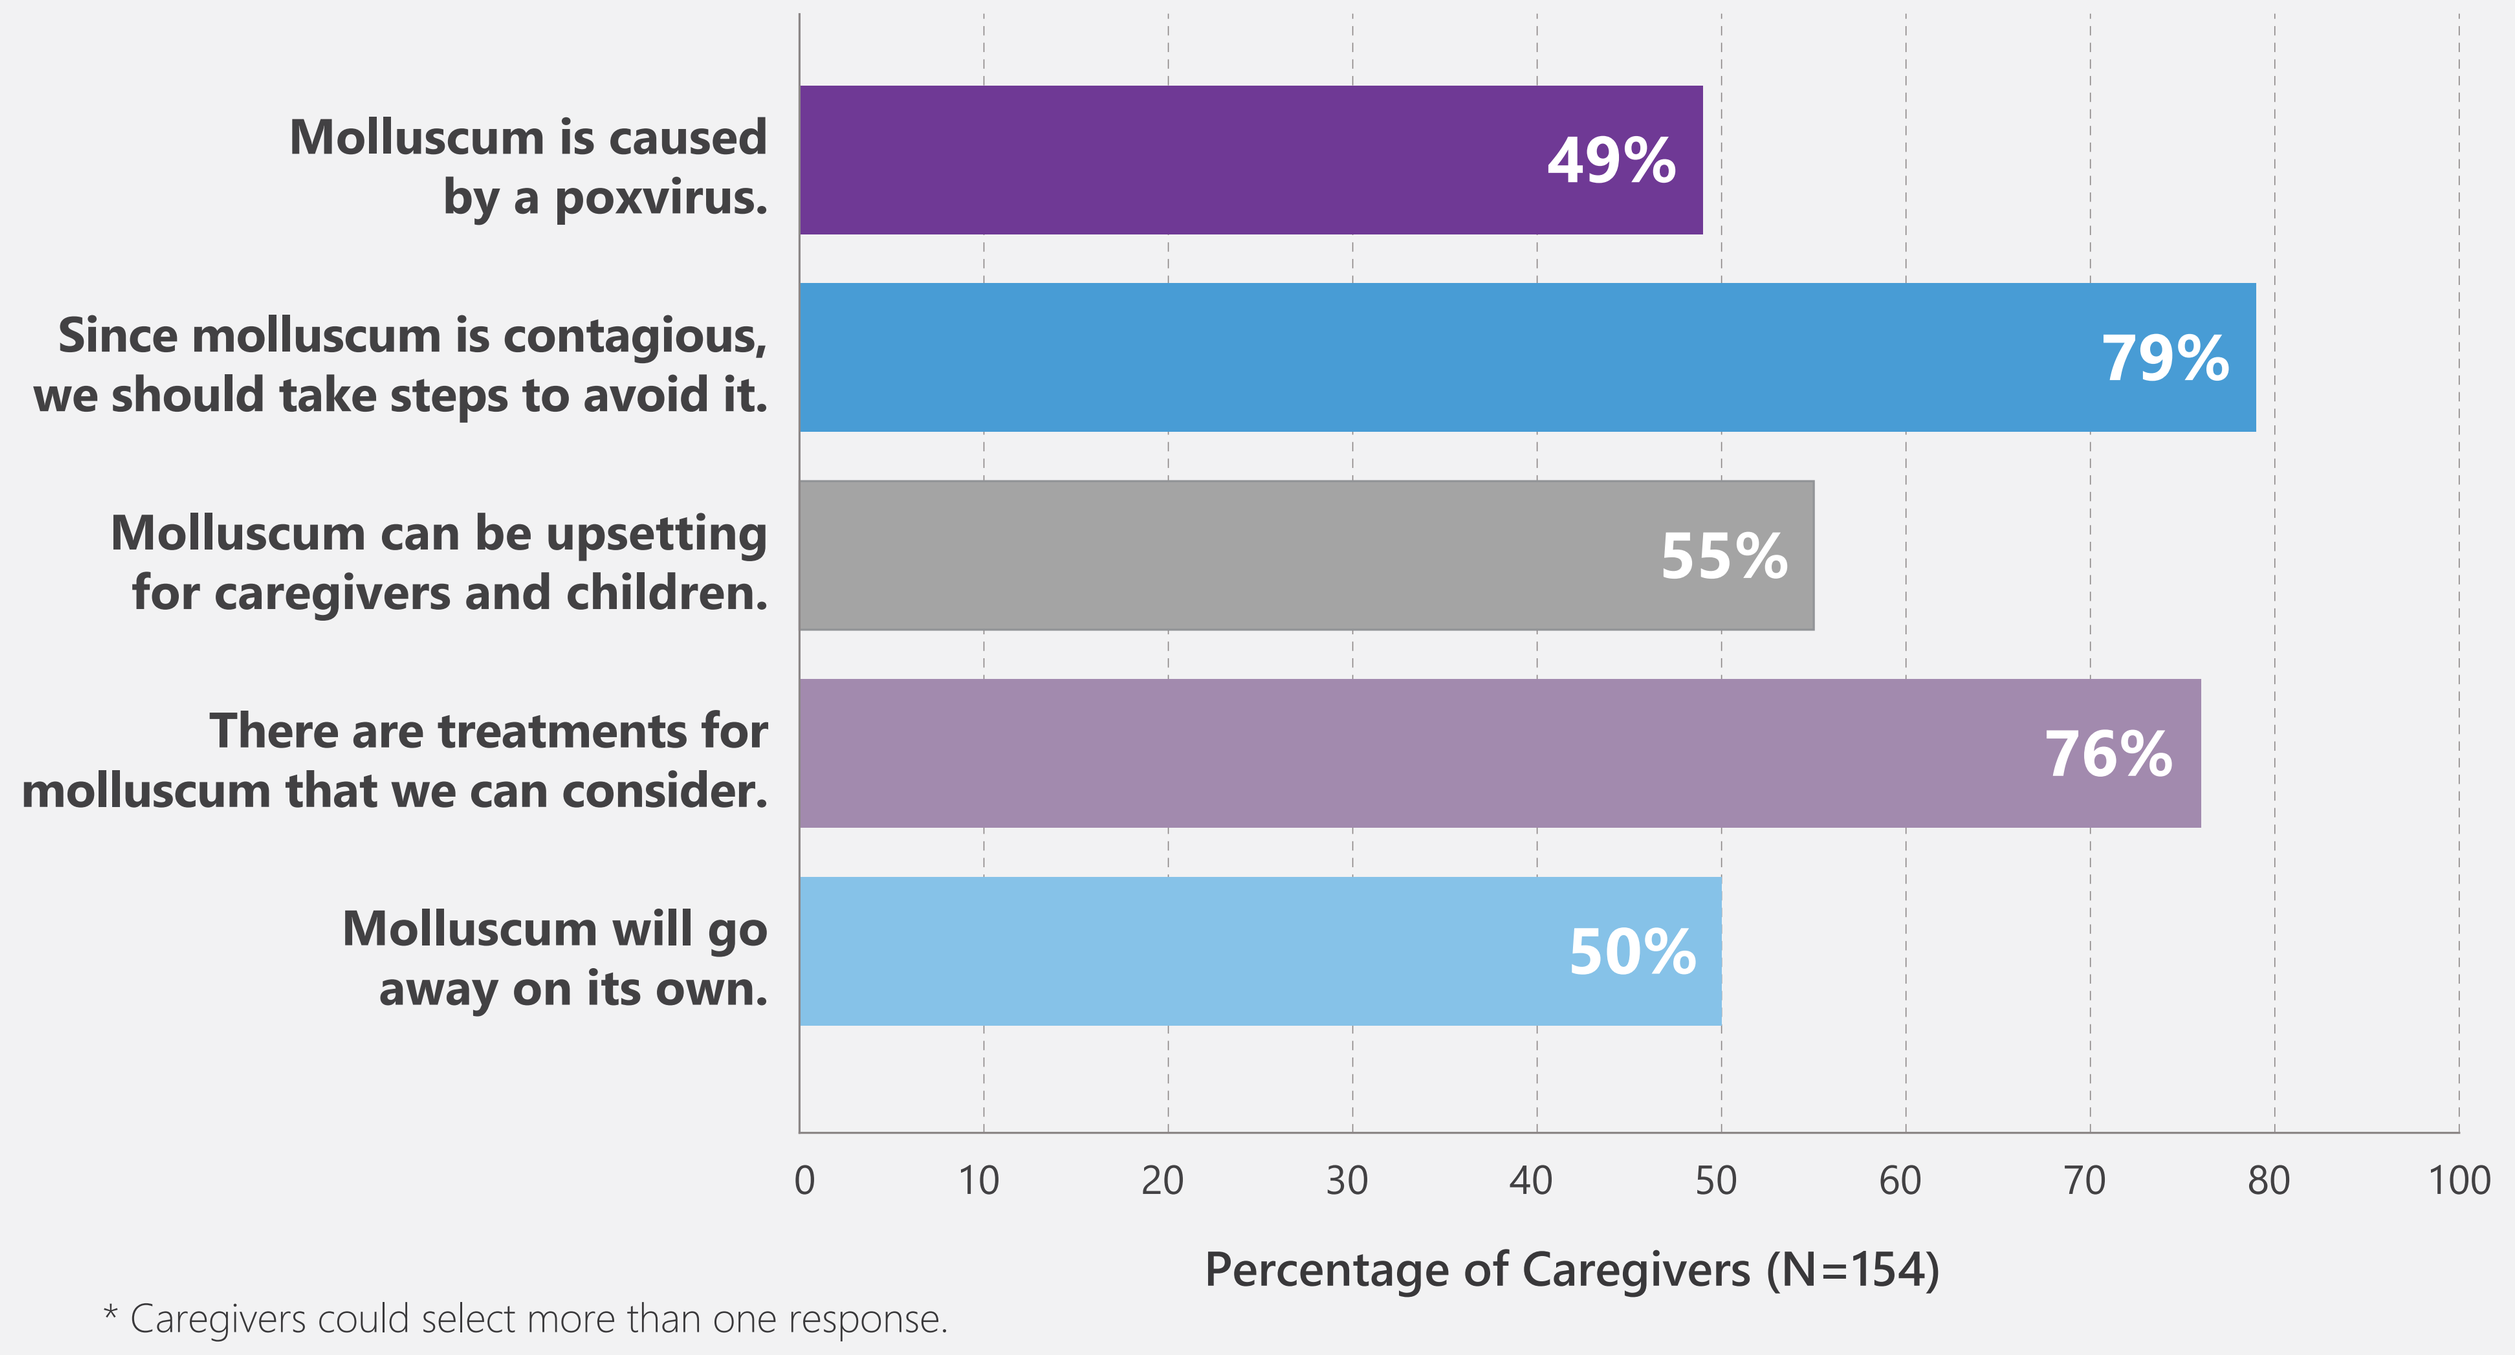
Molluscum is caused by a poxvirus.
49%
Since molluscum is contagious, we should take steps to avoid it.
79%
Molluscum can be upsetting for caregivers and children.
55%
There are treatments for molluscum that we can consider.
76%
Molluscum will go away on its own.
50%
0
10
20
30
40
50
60
70
80
100
Percentage of Caregivers (N=154)

* Caregivers could select more than one response.

Figure 5. "How much of an impact has molluscum had on your child's life (right figure) or on your life (left figure)?

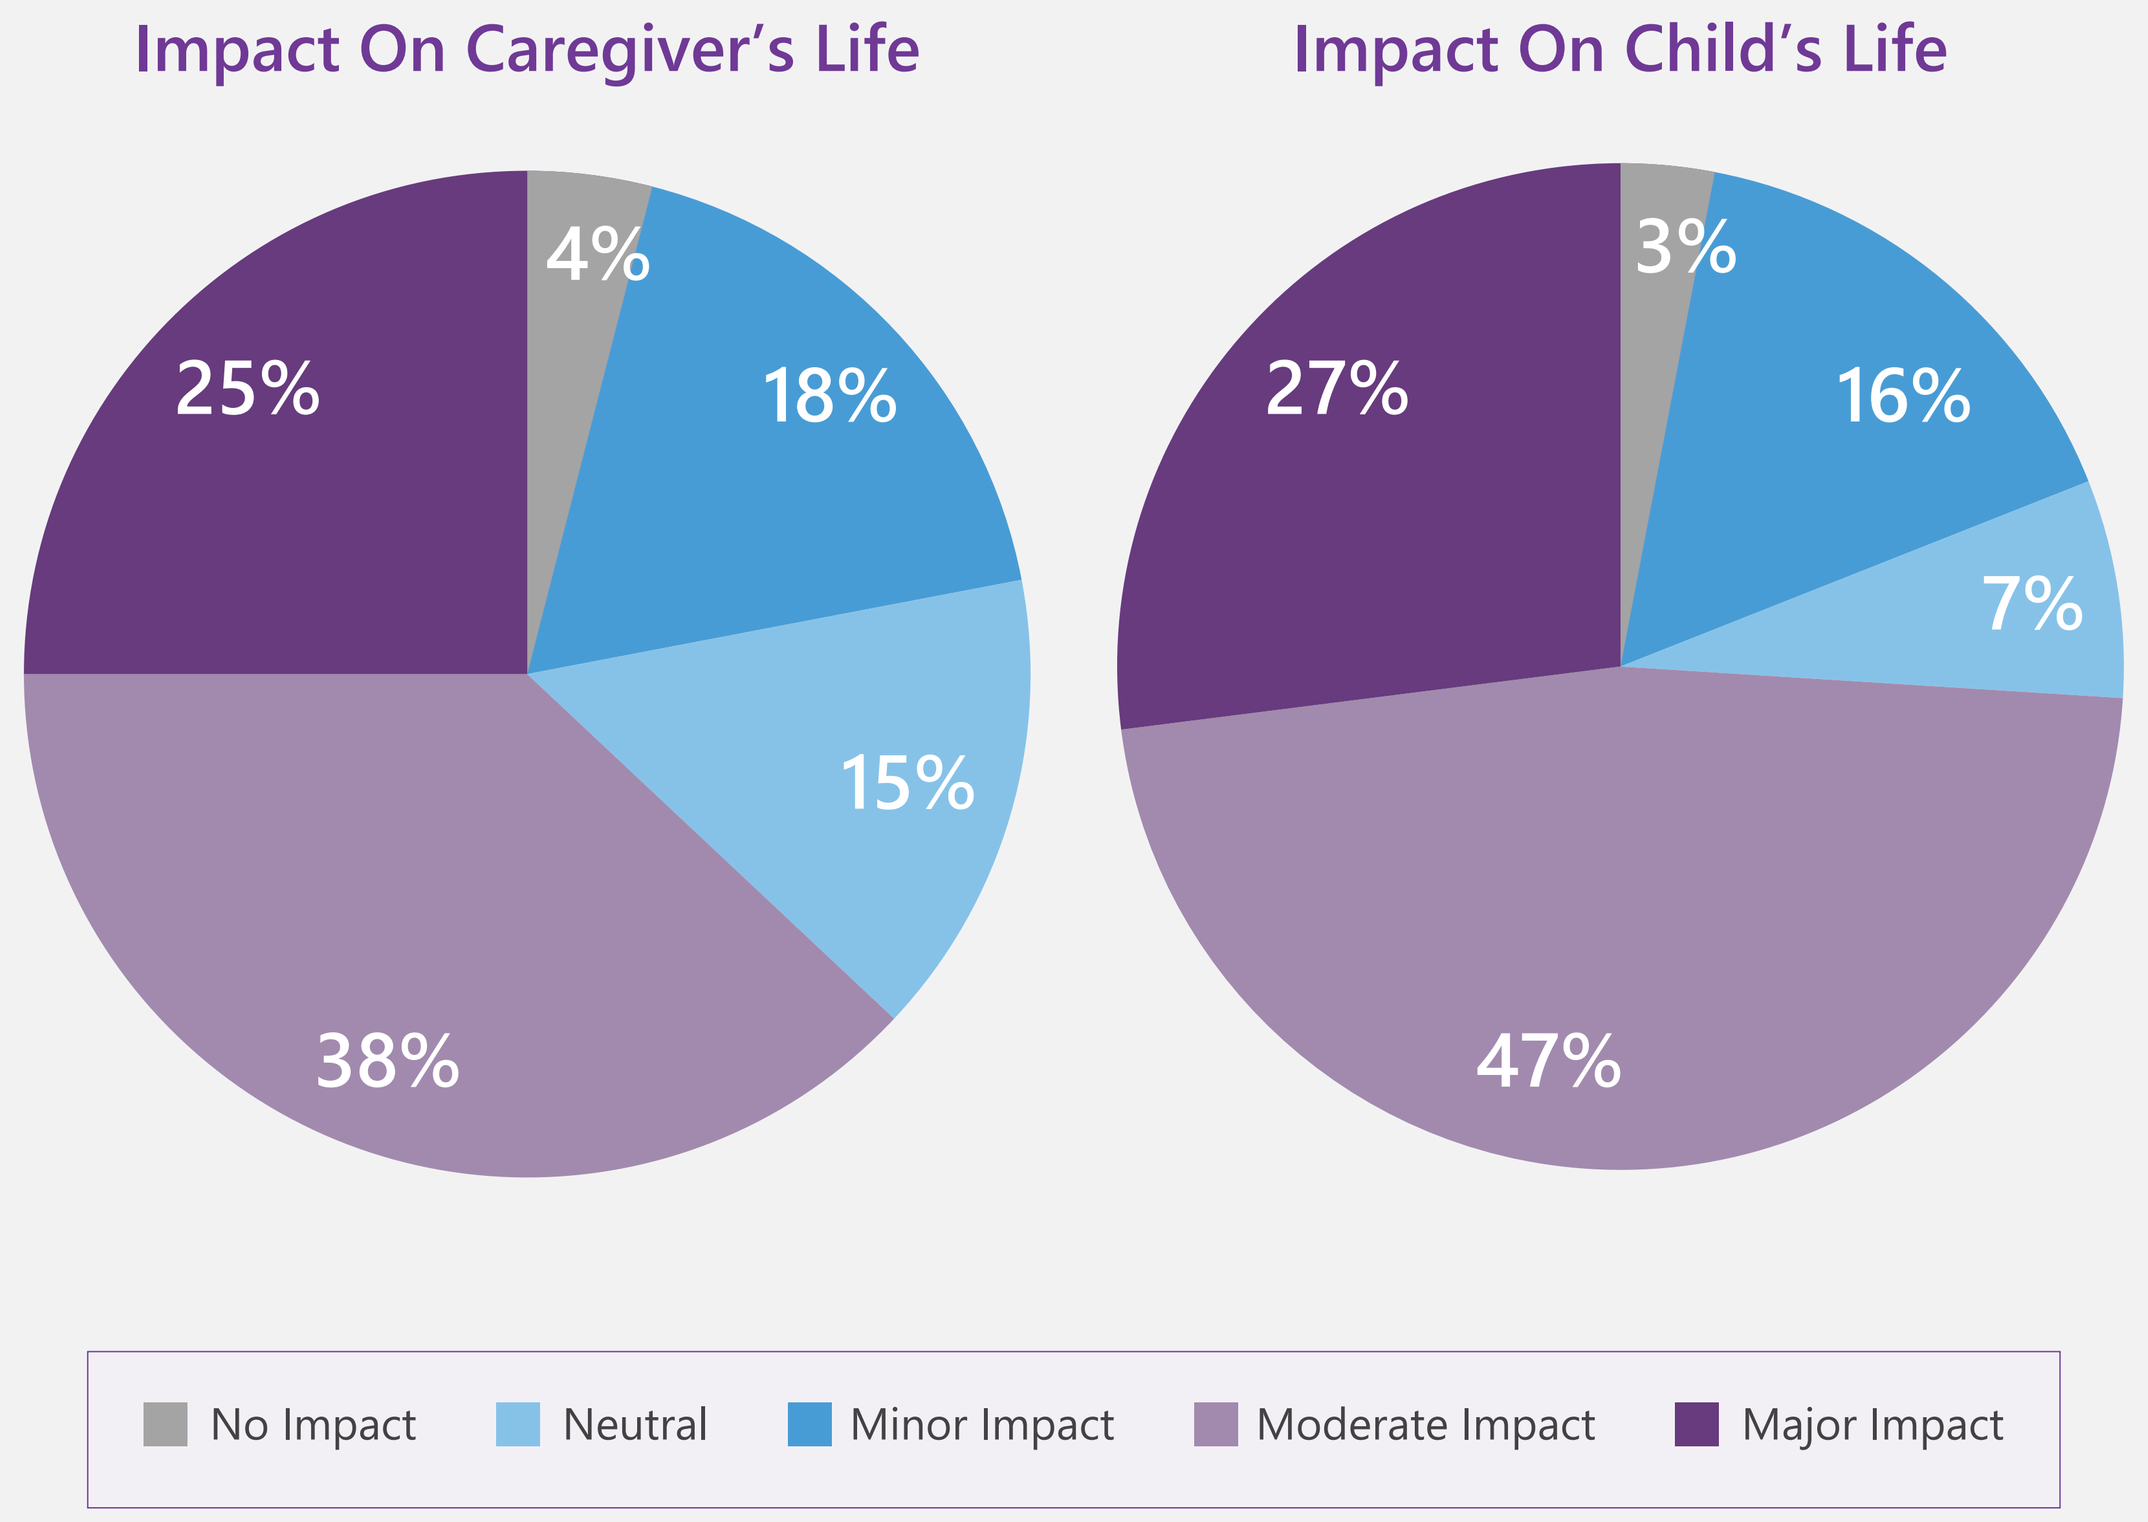
Impact On Caregiver's Life
4%
18%
15%
38%
25%
Impact On Child's Life
3%
16%
7%
47%
27%
No Impact
Neutral
Minor Impact
Moderate Impact
Major Impact

Figure 6. "Which of the following ways of dealing with molluscum did you decide on for your child?"

The average number of treatments tried by caregivers was 2.36.

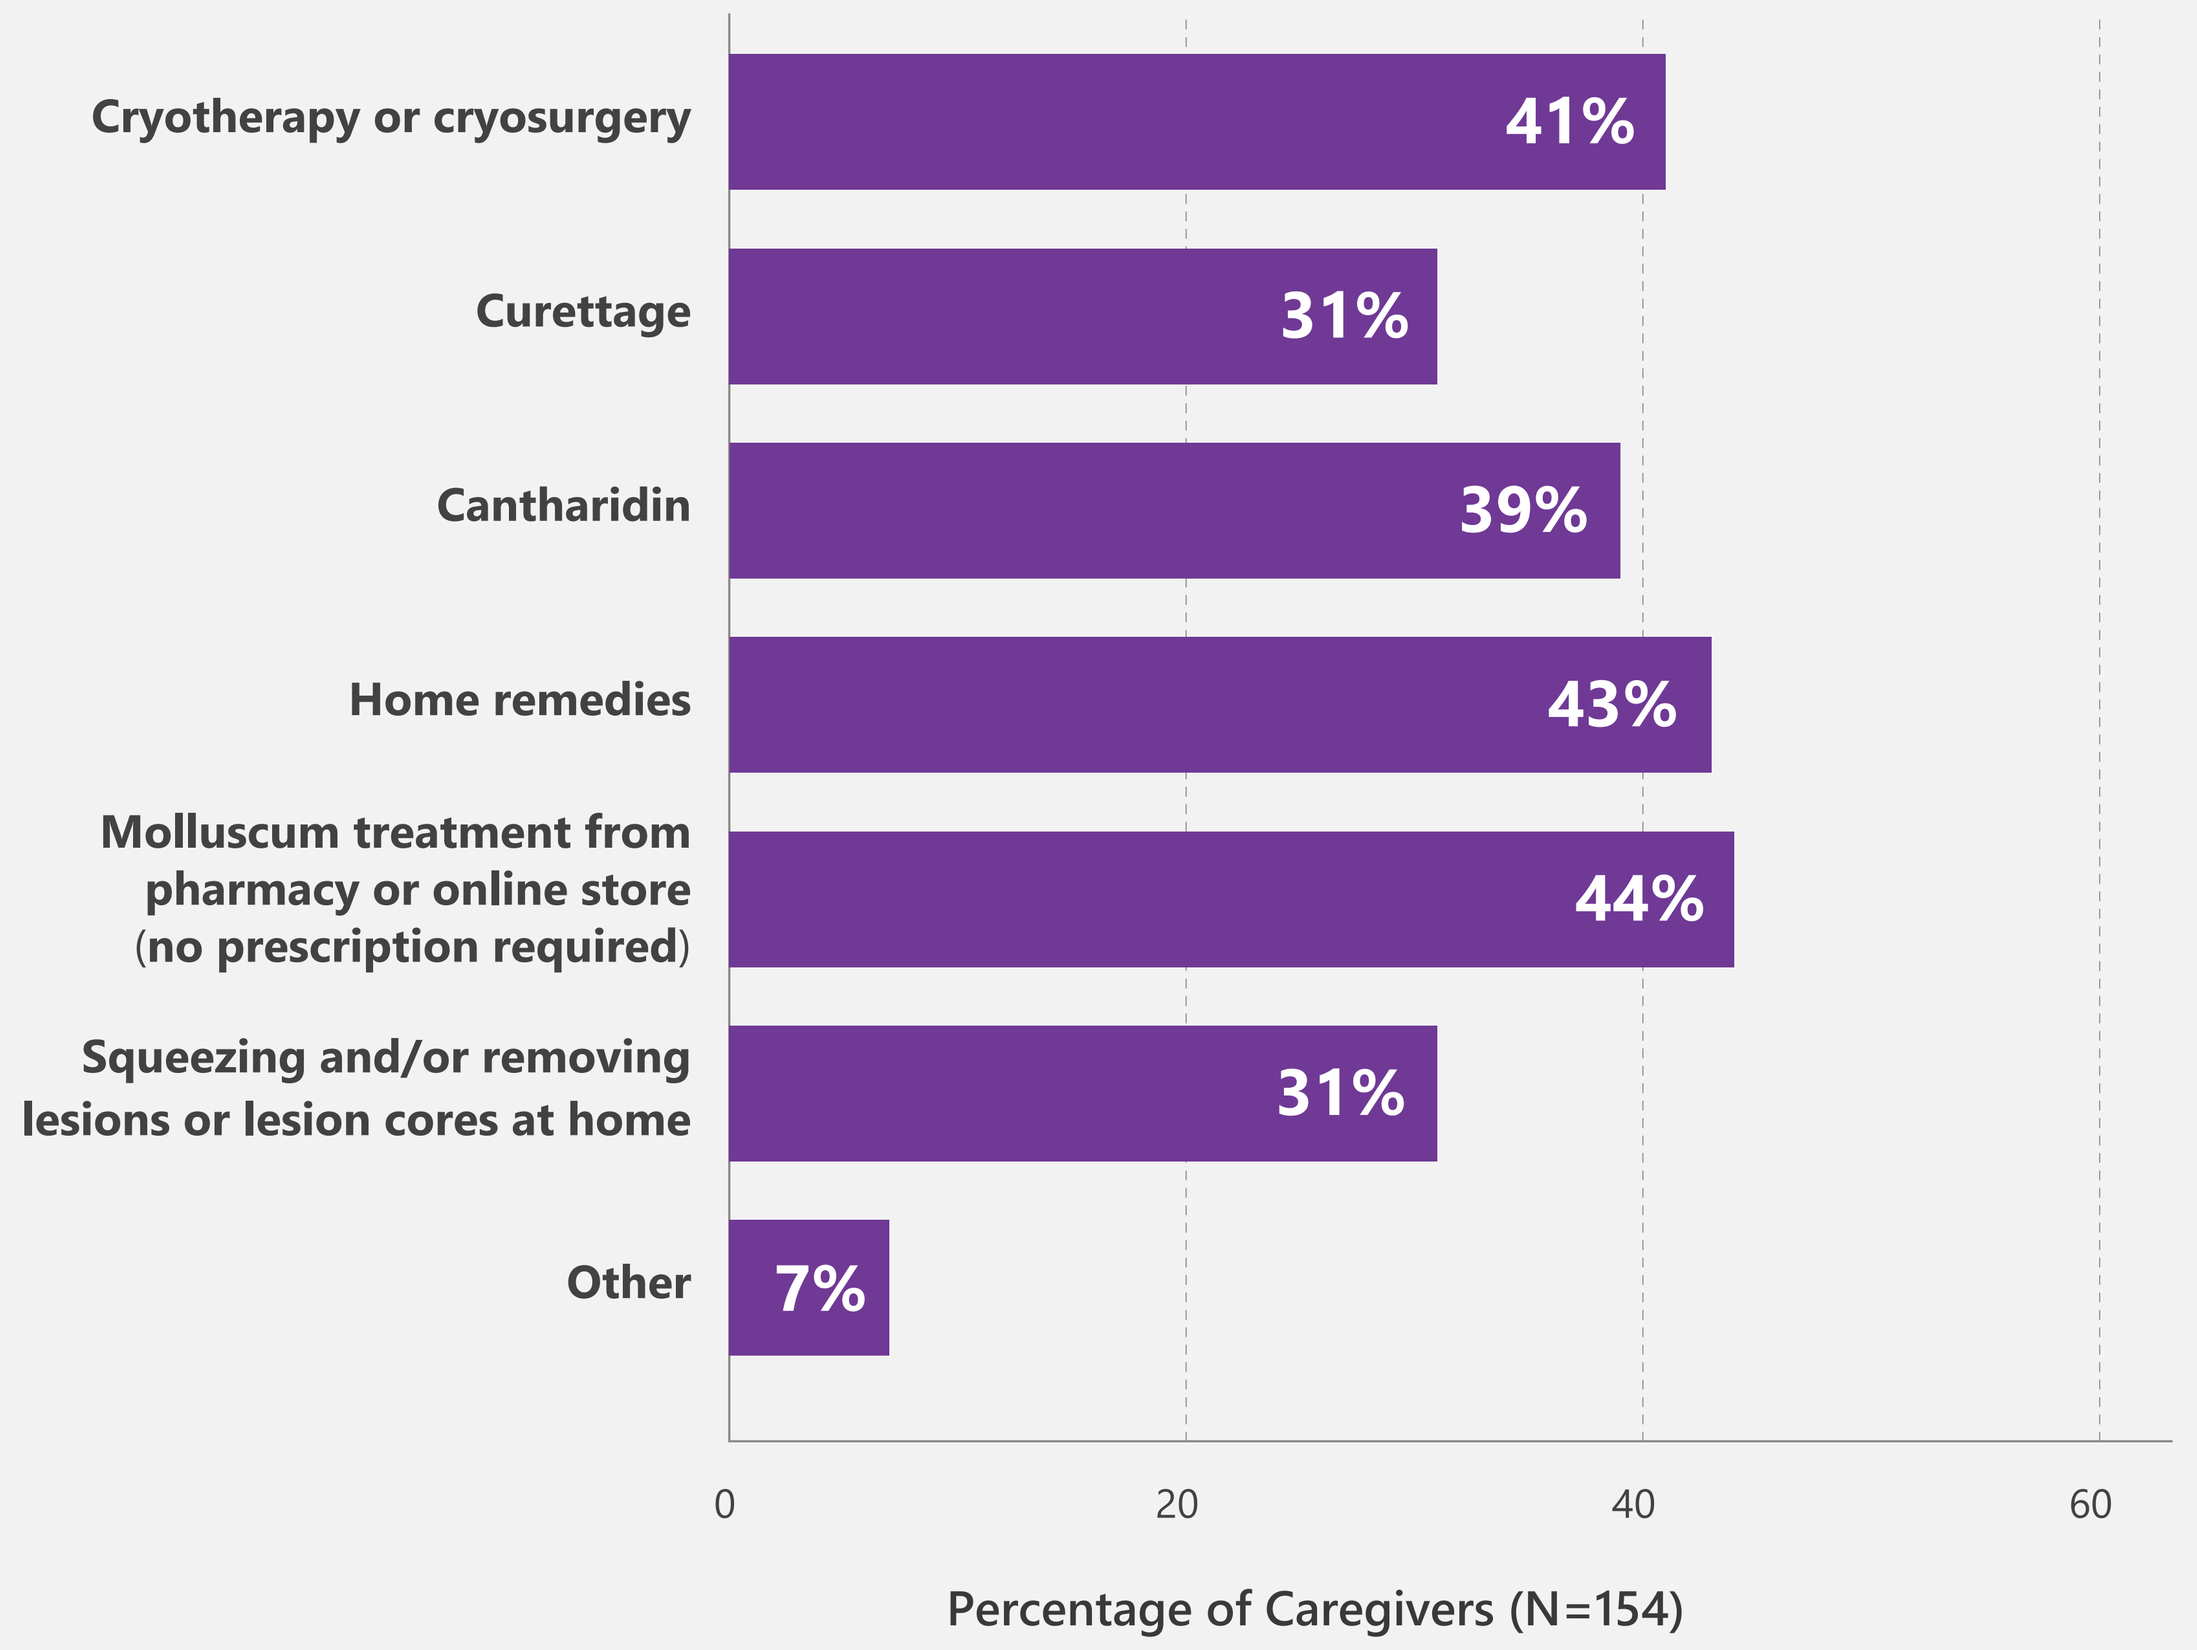
Cryotherapy or cryosurgery
41%
Curettage
31%
Cantharidin
39%
Home remedies
43%
Molluscum treatment from pharmacy or online store (no prescription required)
44%
Squeezing and/or removing lesions or lesion cores at home
31%
Other
7%
0
20
40
60
Percentage of Caregivers (N=154)

CONCLUSIONS

- These survey results indicate that patients with molluscum receive a clinical diagnosis from numerous sources and that many molluscum patients do not receive treatment.
- Caregivers may use more than one treatment modality to rid their children of the infection and are likely to treat using at-home remedies, including physical removal of lesions.
- Spread to another child in the household was common despite being told about the infectious nature of the virus.
- Caregivers reported a substantial impact on their quality of life and on the activities of their children with molluscum.
- There is an unmet need for education of HCPs regarding the diagnosis and impact of molluscum, in addition to safe and effective treatment options for the infection.

Disclosures

The studies were sponsored by Verrica Pharmaceuticals Inc. Editorial support was provided by Versant Learning Solutions and funded by Verrica Pharmaceuticals Inc. The authors have received the following from Verrica Pharmaceuticals:
P. Kwong: H, C; **A. Herbert**: H. **C. Utley**: H; **M. Olivadoti**: E.
H=honoraria; C=clinical funds; S=stocks; E=employee.