Safety and Efficacy of VP-102 in Molluscum Contagiosum by Age Group: Pooled Results of Two Phase 3 Multicenter, Randomized, Vehicle-Controlled Clinical Trials

Elaine Siegfried[1], Scott Katz[2], Pieter d'Arnaud[3], Melissa Olivadoti[4]
[1]St. Louis University, St. Louis, MO; [2]Plano Pediatrics, Plano, TX; [3]Instat Consulting, Inc., Chatham, NJ. [4]Verrica Pharmaceuticals Inc, West Chester, PA.

VP-102

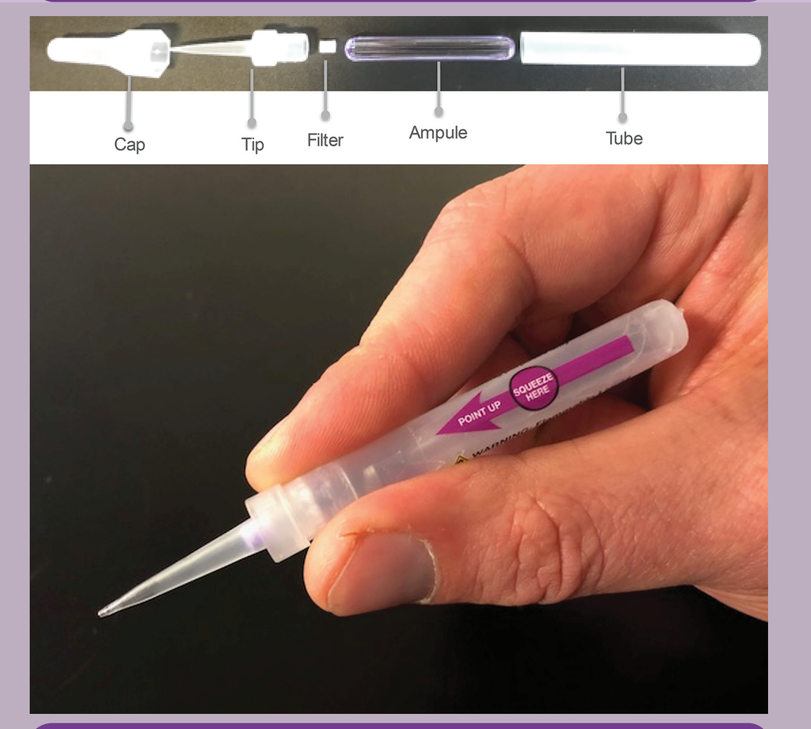
Cap
Tip
Filter
Ampule
Tube

Proprietary drug-device combination product that is not approved by the FDA. Photo is for representative purposes only.

BACKGROUND

- Molluscum contagiosum (molluscum) is a common viral skin infection that predominately affects children.
- VP-102 is a proprietary drug-device combination product containing cantharidin (0.7%, w/v) under investigation for the treatment of molluscum in participants 2 years of age or older.
- These exploratory analyses examined the pooled safety and efficacy data from two Phase 3 clinical trials of VP-102 for treatment of molluscum, stratified by participant baseline age.

METHODS

- Participants ≥ 2 years of age with molluscum were enrolled in two identical Phase 3 trials and randomized in a 3:2 ratio to receive either topical VP-102 or vehicle.
- Study drug was applied topically to all baseline and new lesions once every 21 days until complete clearance of lesions, or up to a maximum of 4 applications.
- Efficacy and safety assessed in pre-specified analyses by age group (2–5y, 6–11y, 12–18y, ≥19y) including:
 - Percentage of participants with complete clearance of all baseline and new lesions by day 84 (end of study visit).
 - Percent change in lesion count from baseline to each visit (days 21, 42, 63, and 84).
 - Incidence of treatment-related treatment emergent adverse events (TR-TEAEs) with a specific focus on related local skin reactions.

RESULTS

Baseline Demographics and Molluscum Medical Histories by Age Group

	VP-102				Vehicle			
	2–5 years (n=137)	6–11 years (n=140)	12–18 years (n=22)	≥19 years (n=11)	2–5 years (n=106)	6–11 years (n=89)	12–18 years (n=18)	≥19 years (n=5)
Age (years)								
Mean (SD)	3.7 (1.04)	8.0 (1.64)	13.2 (1.85)	35.4 (14.4)	3.6 (1.08)	7.7 (1.44)	13.2 (1.66)	37.2 (10.6)
Gender - No. (%)								
Female	64 (46.7)	70 (50.0)	11 (50.0)	9 (81.8)	54 (50.9)	40 (44.9)	10 (55.6)	3 (60.0)
Male	73 (53.3)	70 (50.0)	11 (50.0)	2 (18.2)	52 (49.1)	49 (55.1)	8 (44.4)	2 (40.0)
Ethnicity - No. (%)								
Hispanic/Latino	26 (19.0)	25 (17.9)	4 (18.2)	3 (27.3)	10 (9.4)	14 (15.7)	5 (27.8)	2 (40.0)
Not Hispanic/Latino	111 (81.0)	115 (82.1)	18 (81.8)	8 (72.7)	95 (89.6)	75 (84.3)	13 (72.2)	2 (40.0)
Not Reported	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (0.9)	0 (0.0)	0 (0.0)	1 (20.0)
Race - No. (%)								
American Indian/Alaskan Native	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)	1 (0.9)	0 (0.0)	0 (0.0)	0 (0.0)
Asian	2 (1.5)	2 (1.4)	1 (4.5)	1 (9.1)	0 (0.0)	0 (0.0)	1 (5.6)	0 (0.0)
Black or African American	5 (3.6)	6 (4.3)	2 (9.1)	0 (0.0)	3 (2.8)	4 (4.5)	1 (5.6)	0 (0.0)
White	125 (91.2)	125 (89.3)	17 (77.3)	10 (90.9)	100 (94.3)	82 (92.1)	15 (83.3)	5 (100.0)
Other	5 (3.6)	7 (5.0)	2 (9.1)	0 (0.0)	2 (1.9)	3 (3.4)	1 (5.6)	0 (0.0)
Baseline Lesion Count*								
Mean (SD)	24.0 (24.5)	17.6 (17.0)	13.4 (18.5)	26.8 (55.7)	25.5 (24.4)	21.7 (21.2)	13.7 (10.1)	3.2 (2.2)
Median (Range)	15.0 (1–107)	11.0 (1–103)	8.0 (1–75)	5.0 (1–184)	17.5 (1–110)	15.0 (1–95)	13.0 (1–36)	2.0 (1–6)
Time Since Clinical Diagnosis (Days)								
Mean (SD)	108.9 (159.5)	133.7 (219.9)	195.5 (307.5)	14 (35.7)	97.8 (139.7)	148.8 (239.9)	206 (260.4)	36 (55.1)
Median (Range)	45.0 (1–1,010)	26.0 (1–1,104)	51.0 (1–1,247)	1.0 (1–120)	22.0 (1–715)	46.0 (1–1,302)	107.5 (1–957)	1.0 (1–127)
Any Previous Treatment for Molluscum								
Yes	38 (27.7)	39 (27.9)	9 (40.9)	3 (27.3)	31 (29.2)	33 (37.1)	6 (33.3)	2 (40.0)
Atopic Dermatitis (AD) - No. (%)								
History or Active AD	30 (21.9)	15 (10.7)	5 (22.7)	0 (0.0)	23 (21.7)	10 (11.2)	2 (11.1)	0 (0.0)
Active AD†	12 (8.8)	8 (5.7)	3 (13.6)	0 (0.0)	13 (12.3)	6 (6.7)	1 (5.6)	0 (0.0)

ITT population.

* The *n* for the baseline lesion count in subjects 6 –11 years was 139 (not 140).

† Active atopic dermatitis was determined by concomitant medication usage of the following medications during the study: topical corticosteroids, topical calcineurin inhibitors, and phosphodiesterase-4 inhibitors.

Percentage of Participants With Complete Clearance by Age Group and Timepoint

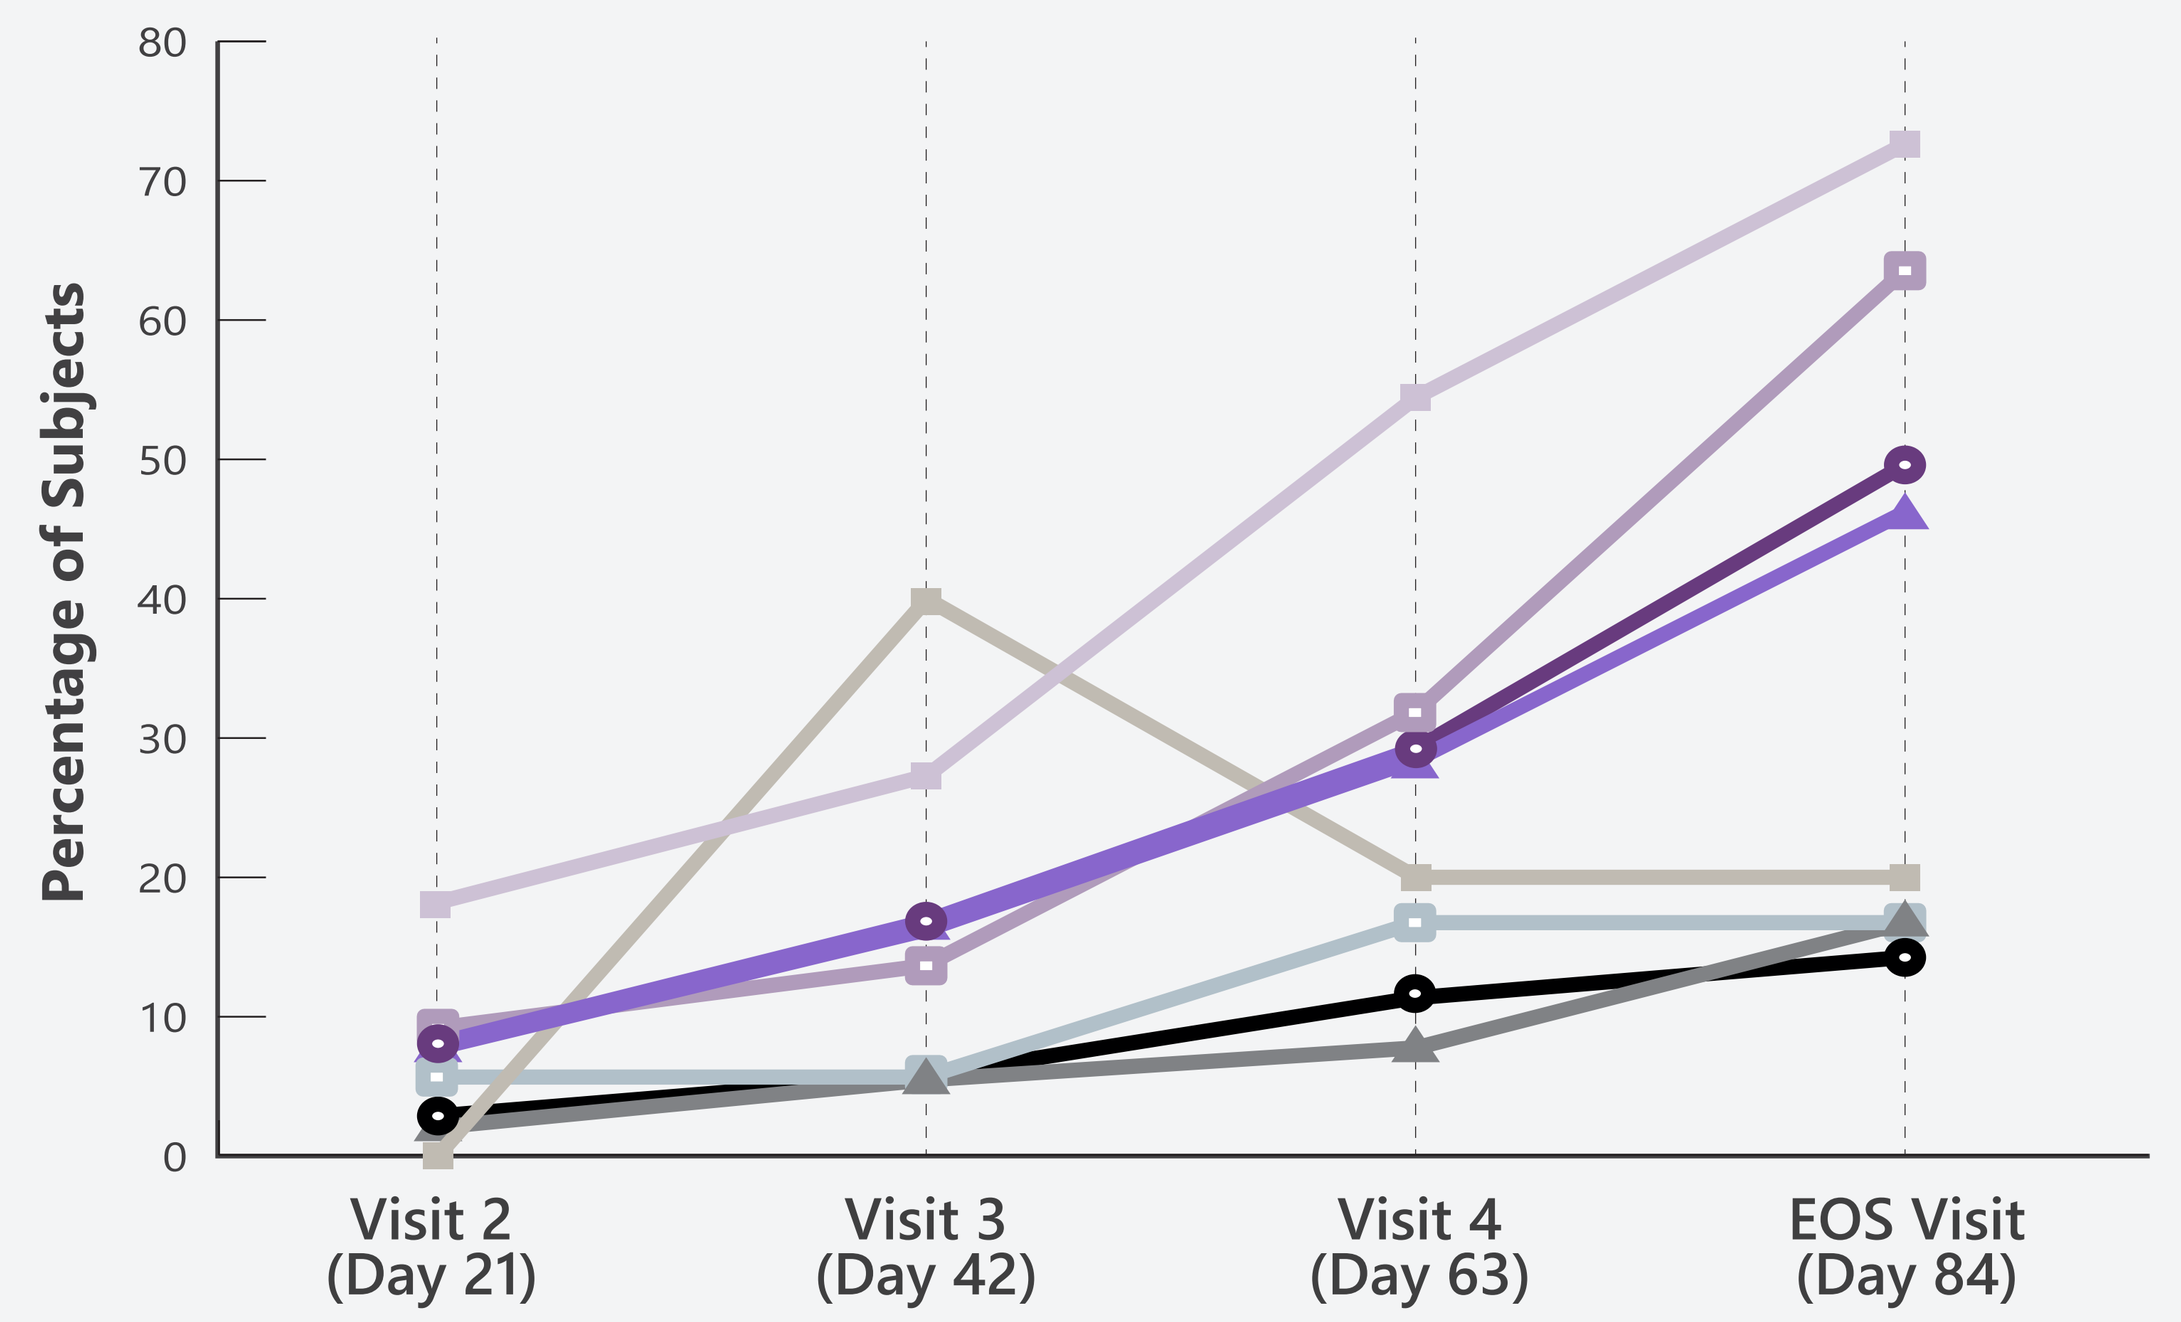
Percentage of Subjects
0
10
20
30
40
50
60
70
80
Visit 2 (Day 21)
Visit 3 (Day 42)
Visit 4 (Day 63)
EOS Visit (Day 84)

		p value vs. Vehicle			
		Age Group			
		2–5	6–11	12–18	≥19
Visit (Day)	2 (21)	**0.0125**	**0.0172**	0.7250	**0.0487**
	3 (42)	**0.0001**	**0.0010**	0.3496	0.2249
	4 (63)	**<0.0001**	**<0.0001**	0.2122	0.2688
	EOS (84)	**<0.0001**	**<0.0001**	**<0.0001**	**0.0161**

	VP-102	Vehicle
2–5 years	n=137	n=106
6–11 years	n=140	n=89
12–18 years	n=22	n=18
≥19 years	n=11	n=5

ITT population; EOS=end of study.

Total Incidence of TR-TEAES by Age Group in VP-102-Treated Participants

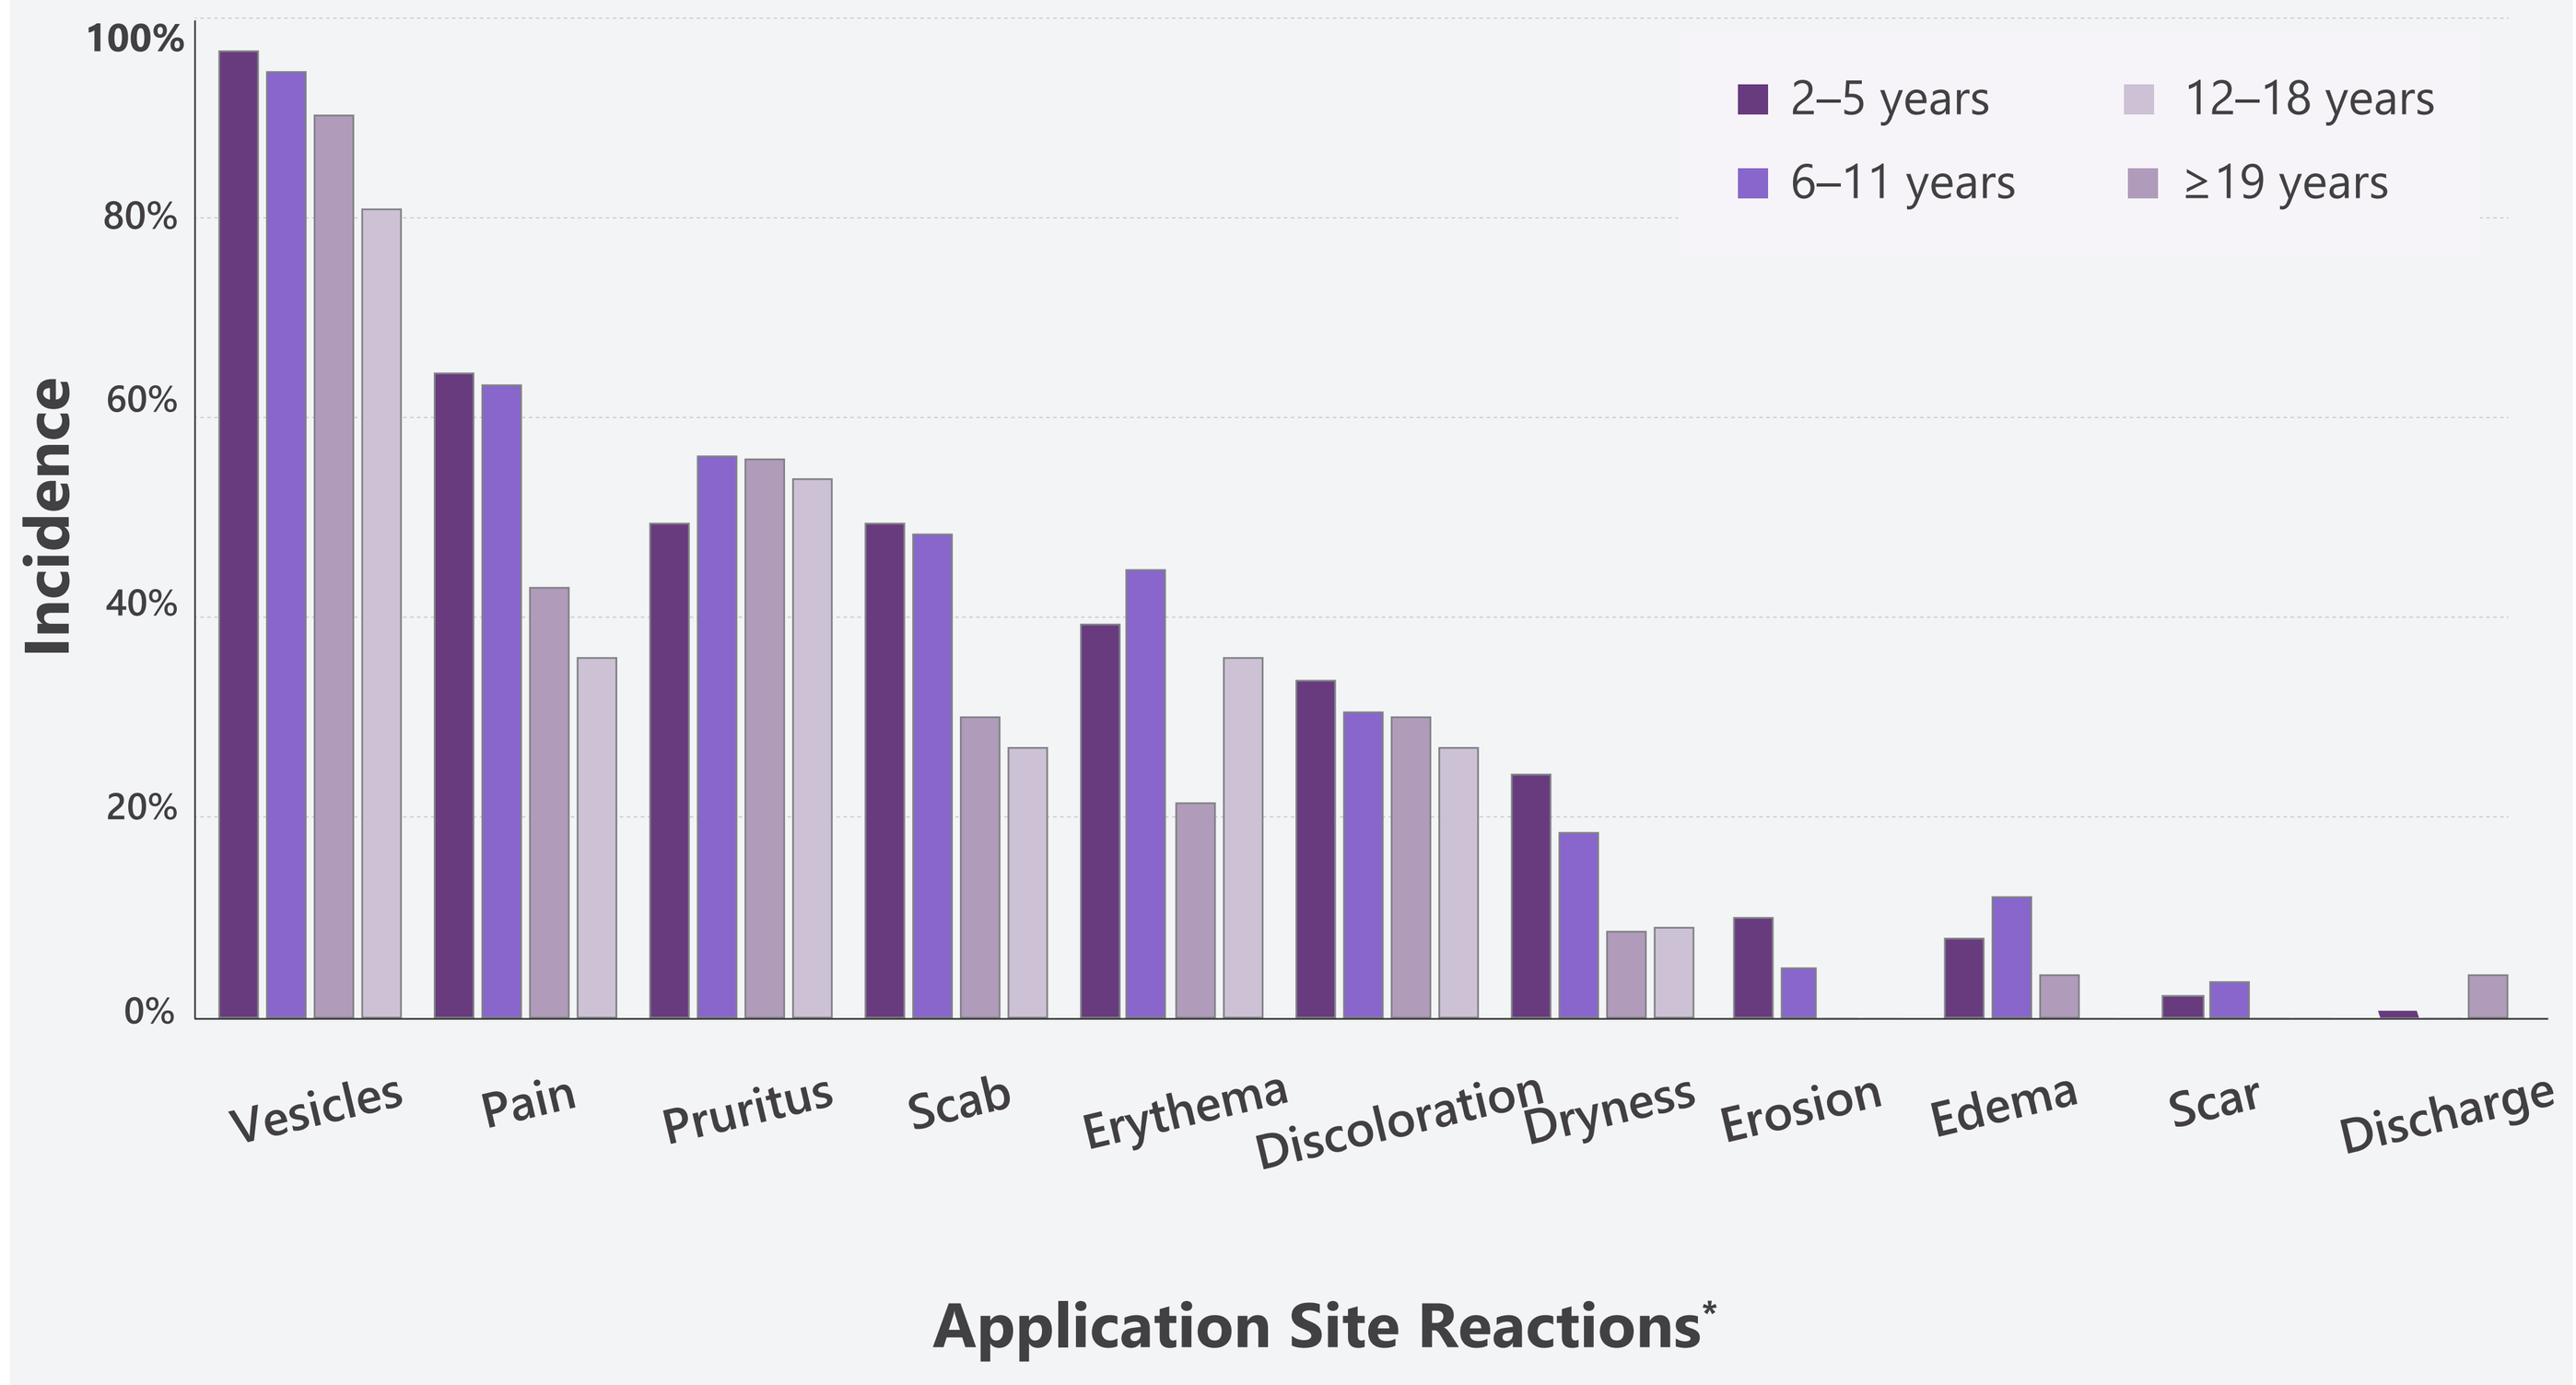
Incidence
0%
20%
40%
60%
80%
100%
2–5 years
6–11 years
12–18 years
≥19 years
Vesicles
Pain
Pruritus
Scab
Erythema
Discoloration
Dryness
Erosion
Edema
Scar
Discharge
Application Site Reactions*

Safety population.

* TR-TEAE reported at an incidence of 2% or greater.

Percent Change from Baseline in Number of Lesions by Age Group at Day 84/EOS

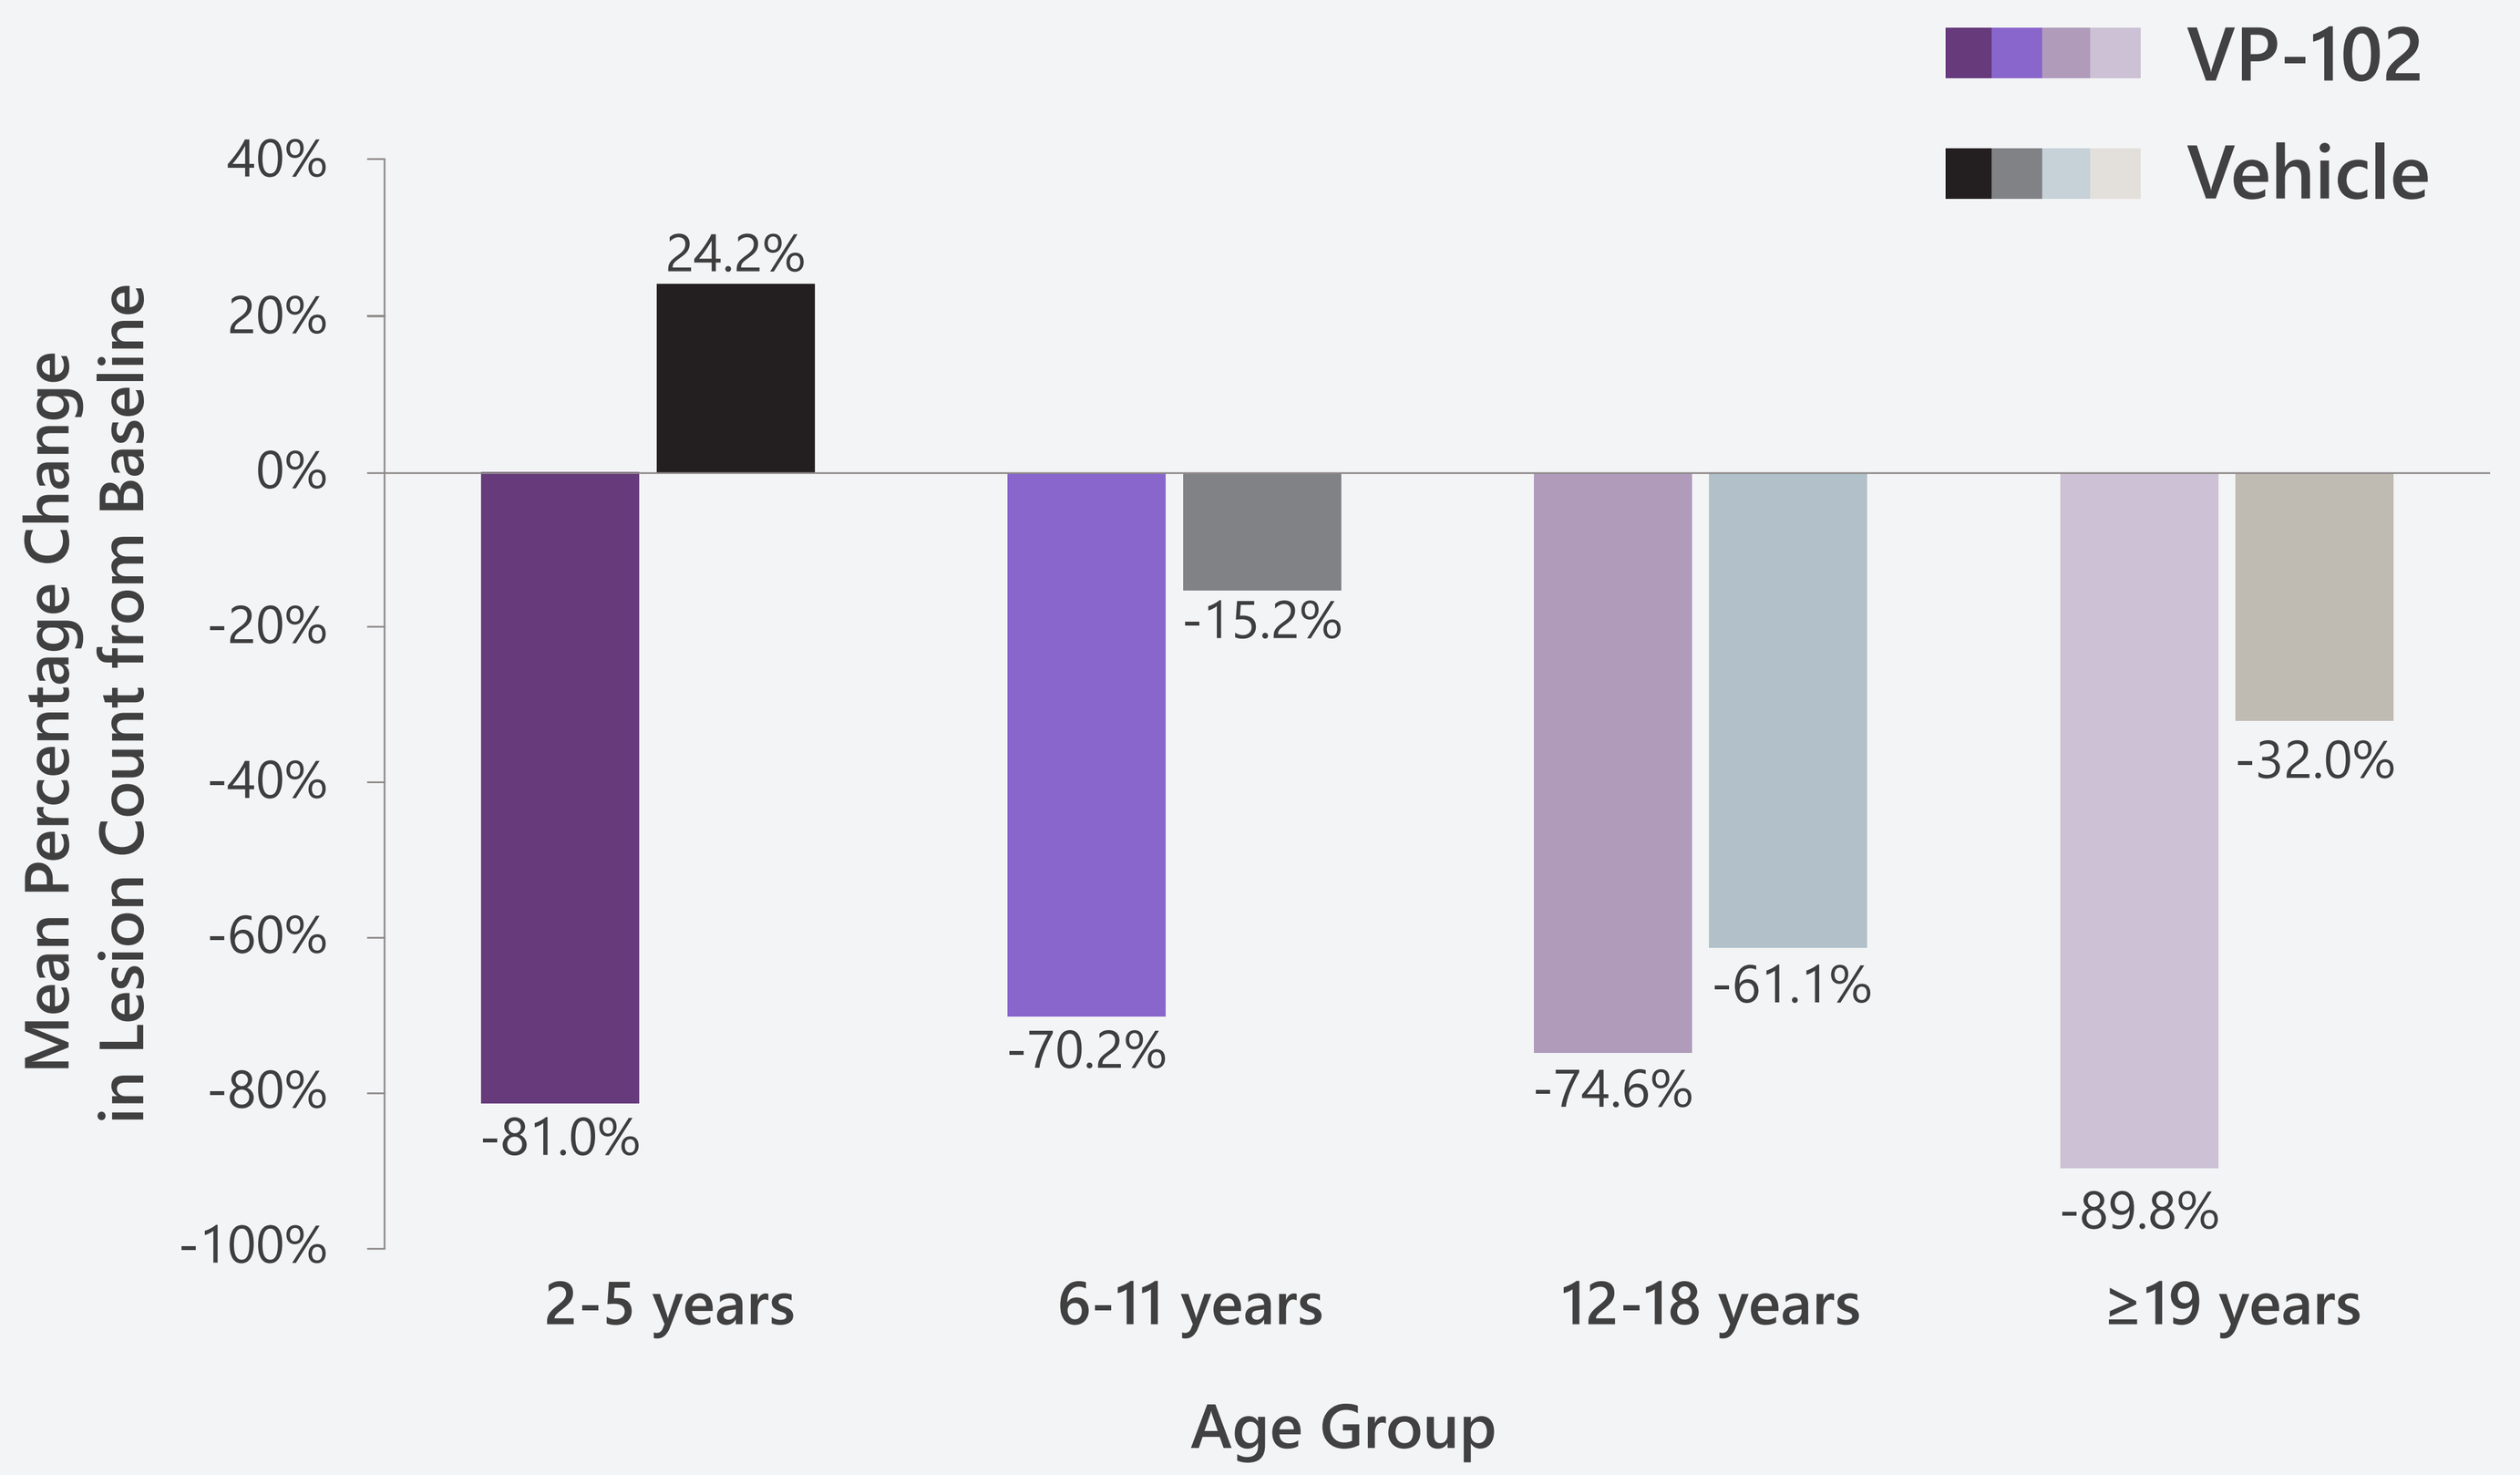
VP-102
Vehicle
Mean Percentage Change in Lesion Count from Baseline
40%
20%
0%
-20%
-40%
-60%
-80%
-100%
24.2%
-81.0%
-70.2%
-15.2%
-74.6%
-61.1%
-89.8%
-32.0%
2-5 years
6-11 years
12-18 years
≥19 years
Age Group

ITT population; EOS=end of study.

CONCLUSIONS

- Baseline lesion count was highest in younger children (ages 2-5), while older children (ages 12-18) and adults were more likely to have received previous treatment.
- Treatment of molluscum with VP-102 showed statistically significantly higher percentage of participants with complete clearance across all age groups compared to vehicle at day 84/EOS, and differences in percent change from baseline in lesion counts.
- TR-TEAEs were mostly mild to moderate in the VP-102 group and incidence was similar across all age groups.
- VP-102 has the potential to be used in all age groups. Due to a limited number of adult participants, further studies are needed to make conclusions in this population.

Disclosures

The studies were sponsored by Verrica Pharmaceuticals Inc. Editorial support was provided by Versant Learning Solutions and funded by Verrica Pharmaceuticals Inc. The authors have received the following from Verrica Pharmaceuticals:
E. Siegfried: H, C; **S. Katz**: H, C; **P. d'Arnaud**: contractor for Verrica; **M. Olivadoti**: E.

H=honoraria; C=clinical funds; S=stocks; E=employee.